Lab of transcriptional memory

Лаборатория транскрипционной памяти

Vera Vecherskaia, Dmytro Fedorenko, Viacheslav Kiselev, Veronika Podliesnova, Sofiya Babok, Anna Kogan

Transcriptional memory - cellular phenomenon of increased rates of gene expression after re-stimulation at a later time

In this lab we analysed microscopy data of wet lab experiments and aimed to use dynamics of transcriptional memory to infer its mechanisms

Транскрипционная память - явление повышенной экспрессии гена в клетке после повторной стимуляции через промежуток времени

В нашей лаборатории мы проанализировали данные экспериментов, чтобы выяснить, каковы механизмы транскрипционной памяти, исходя из её динамики

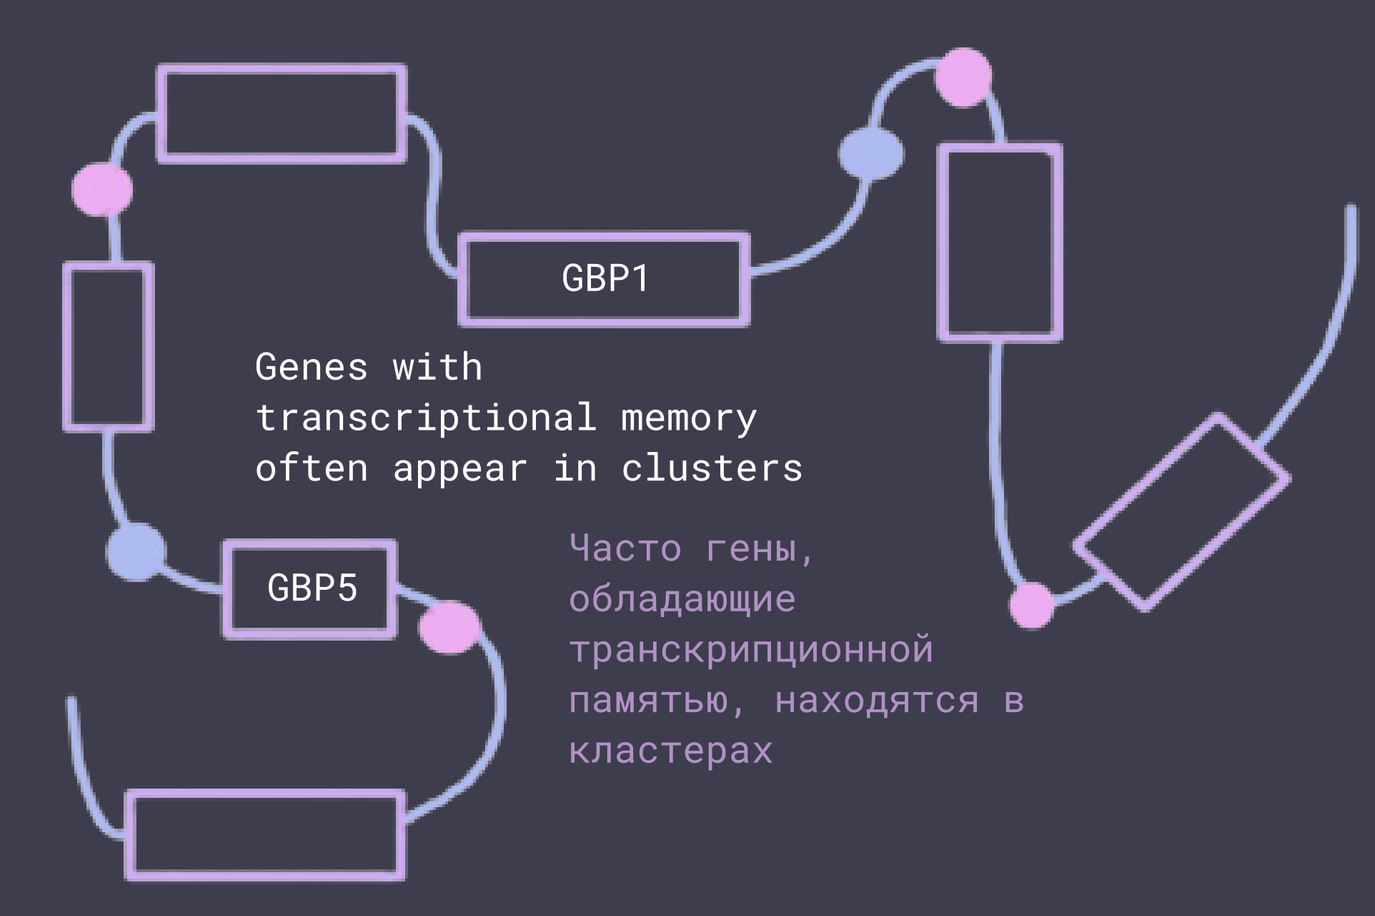
GBP1
GBP5
Genes with transcriptional memory often appear in clusters
Часто гены, обладающие транскрипционной памятью, находятся в кластерах

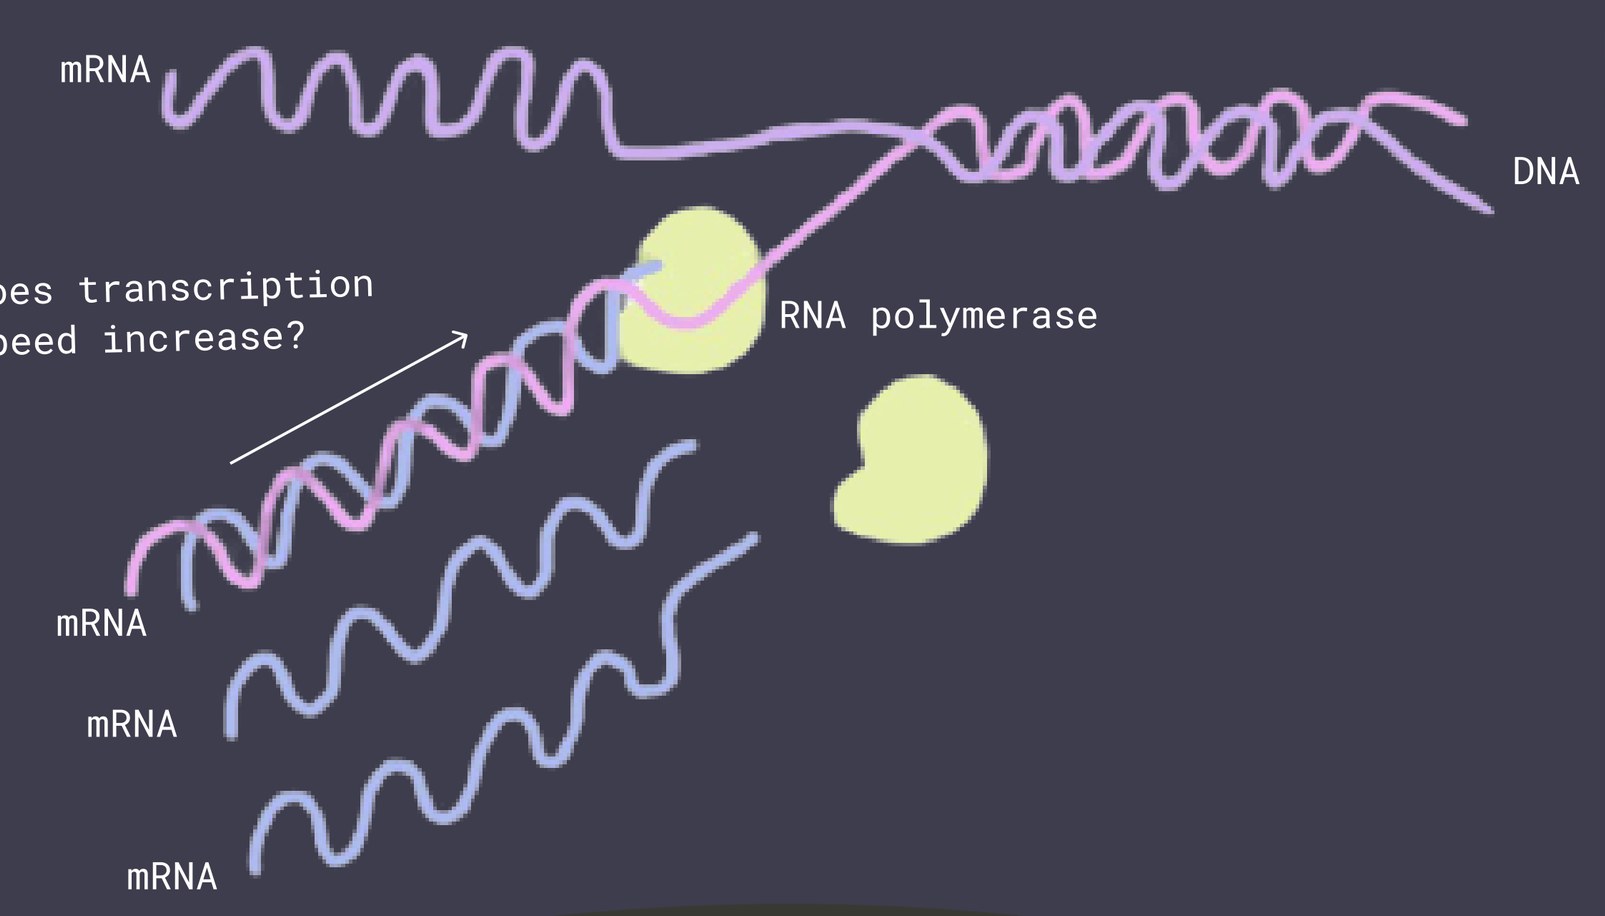
mRNA
DNA
Does transcription speed increase?
RNA polymerase
mRNA
mRNA
mRNA

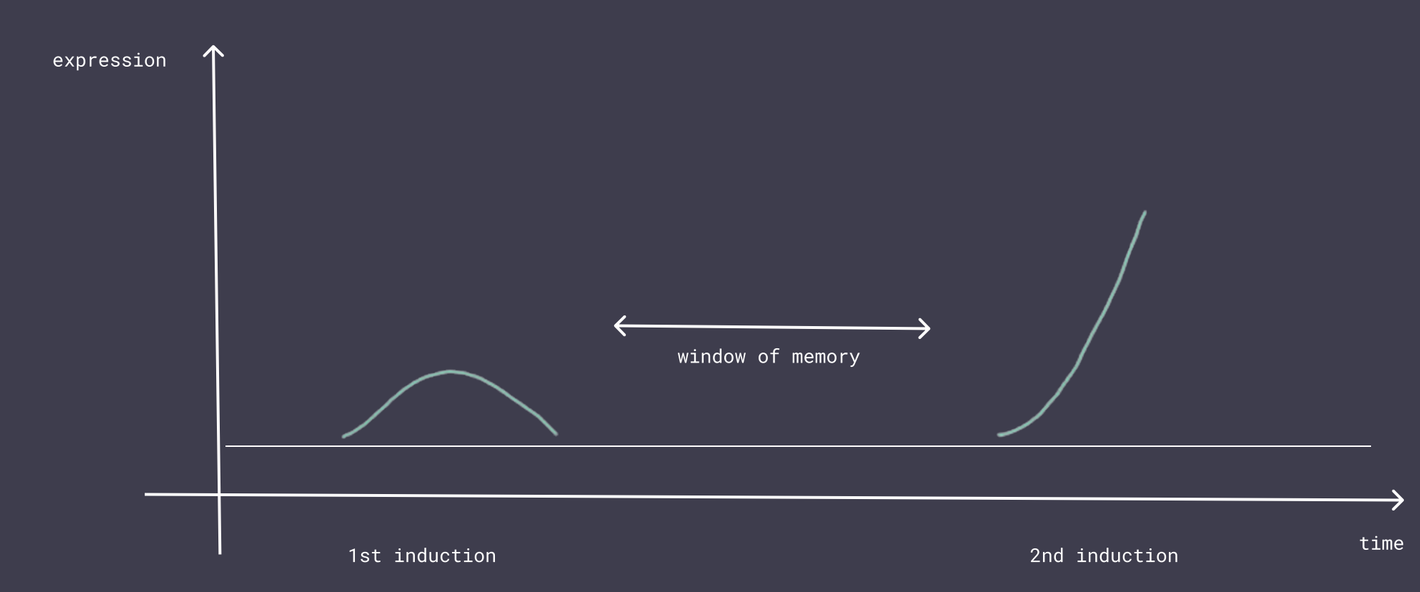
expression
window of memory
1st induction
2nd induction
time

Methods 1//Методы 1
Single-molecule RNA FISH;
Images of fluorescent wide-field microscopy

Software used/
/Использованное программное обеспечение
FIJI
RStudio

We used HeLa cells - some of them are triploid (3n)
Мы использовали раковые клетки HeLa - некоторые из них триплоидны (3n)

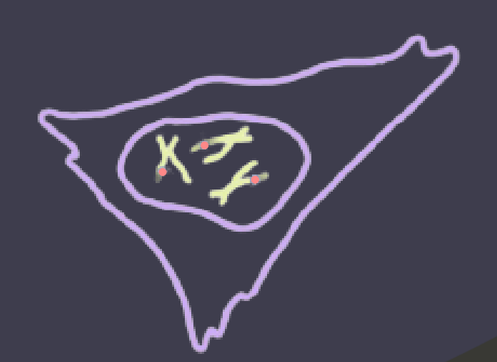

experiment 1 - data for genes GBP5 and GBP1 - first and second responce to INFγ were measured by flourescence

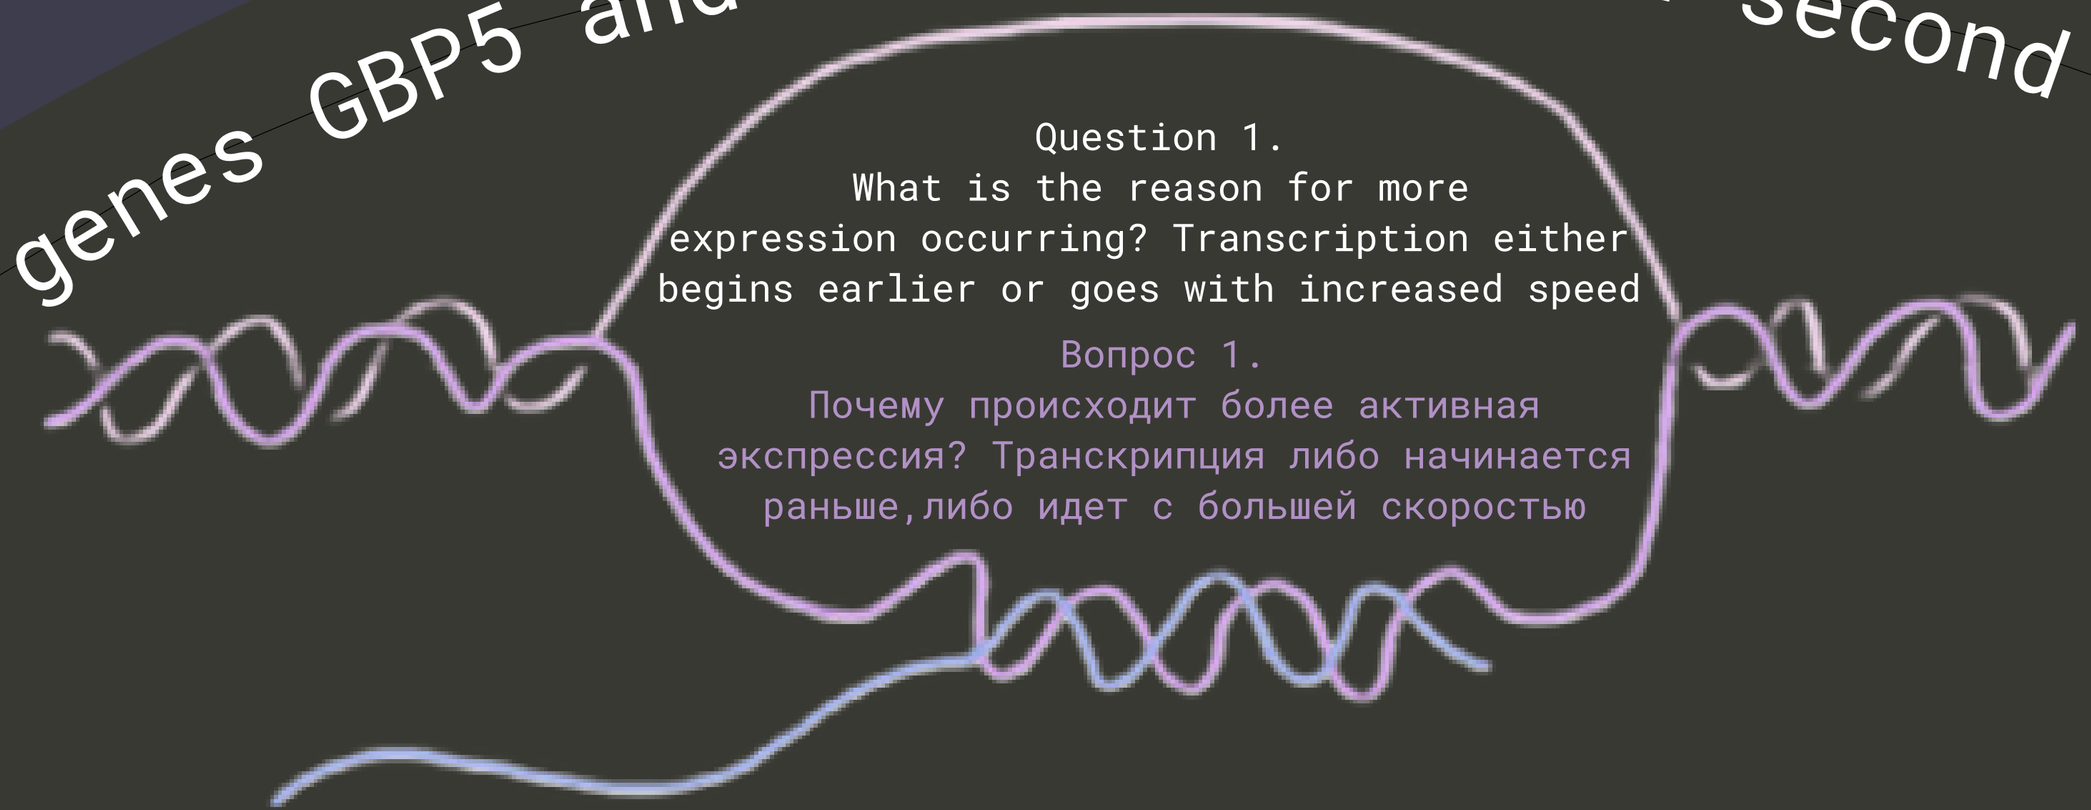
Question 1.
What is the reason for more expression occurring? Transcription either begins earlier or goes with increased speed
Вопрос 1.
Почему происходит более активная экспрессия? Транскрипция либо начинается раньше,либо идет с большей скоростью

GBP5 and GBP1 respectively:
plot 1, plot 4 - weighted histogramms - show, how many transcriptional sites are in a cell right now (usually 0-6); characterises transcriptional bursts

plot 2, plot 5 - dotplots - shows the amount of gene's RNA presented per cell

plot 3, plot 6 - dotplots - shows number of RNAs produced in a cell at a given time point

plot 1

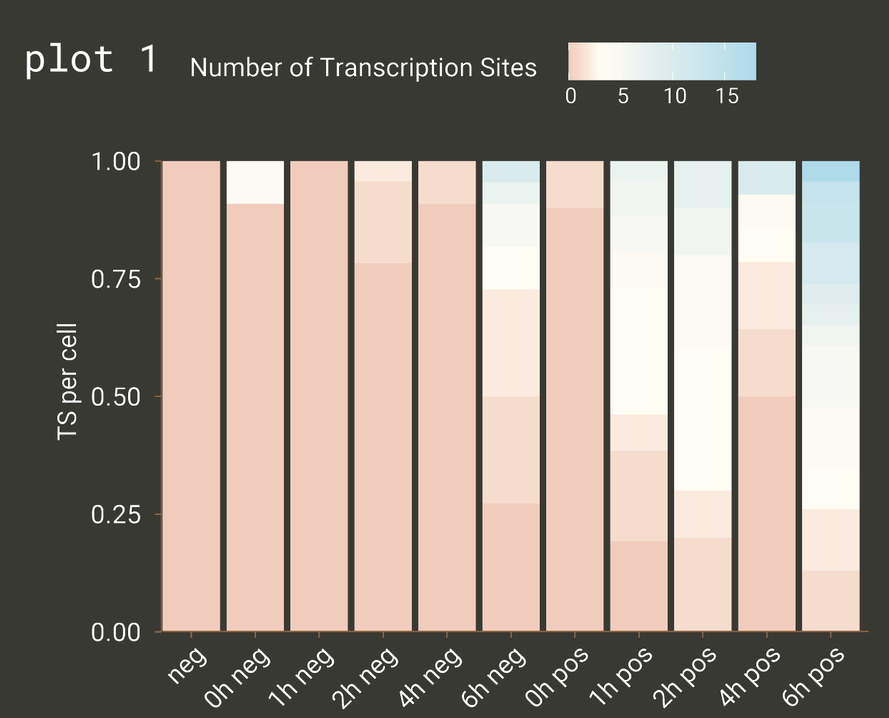
Number of Transcription Sites
TS per cell

Number of GBP5 mRNAs per cell at different time points
plot 2

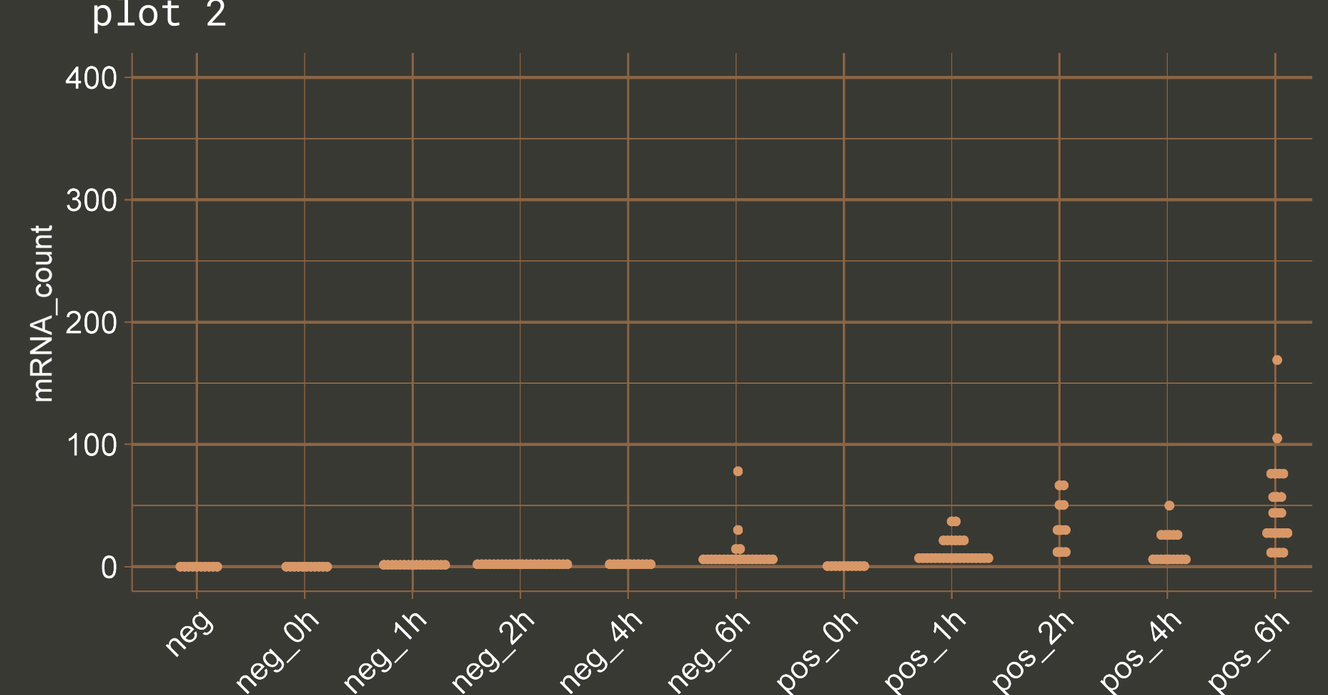
mRNA_count

plot 3
Distribution of TS Integrated densities for GBP5

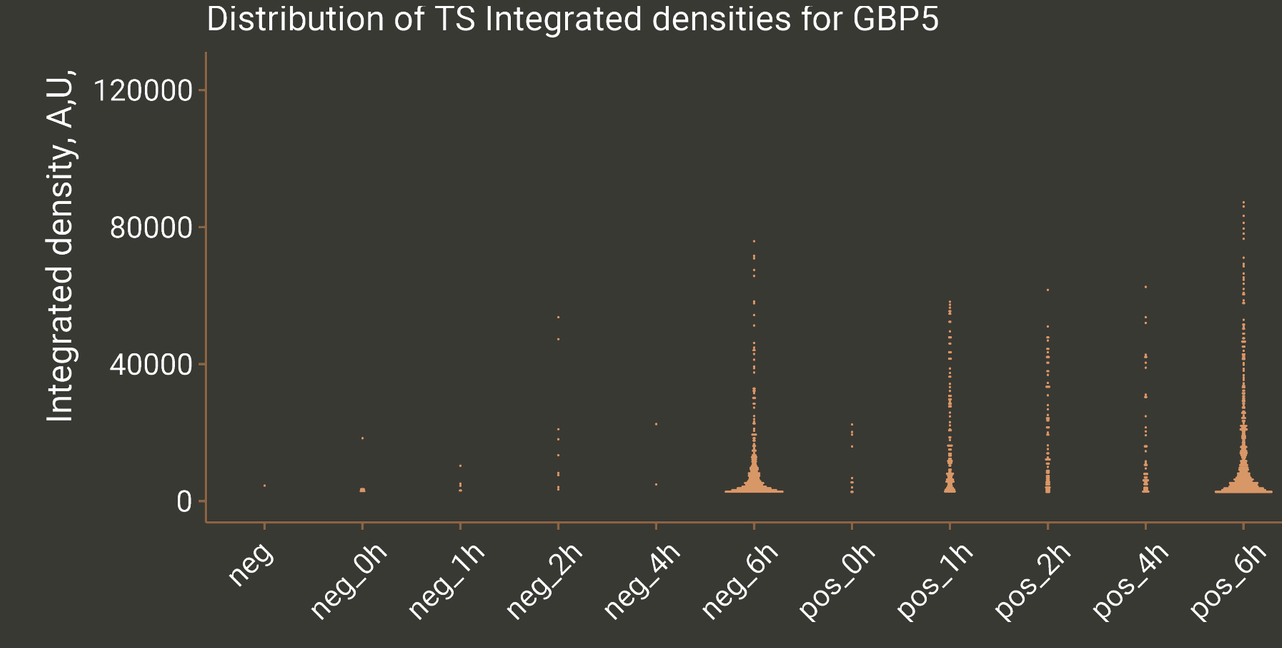
Integrated density, A.U,

GBP5 - transcription occurs earlier, speed can't be compared

По результатам GBP5 нельзя сделать вывод о скорости транскрипции, но транскрипция начинается раньше

plot 4

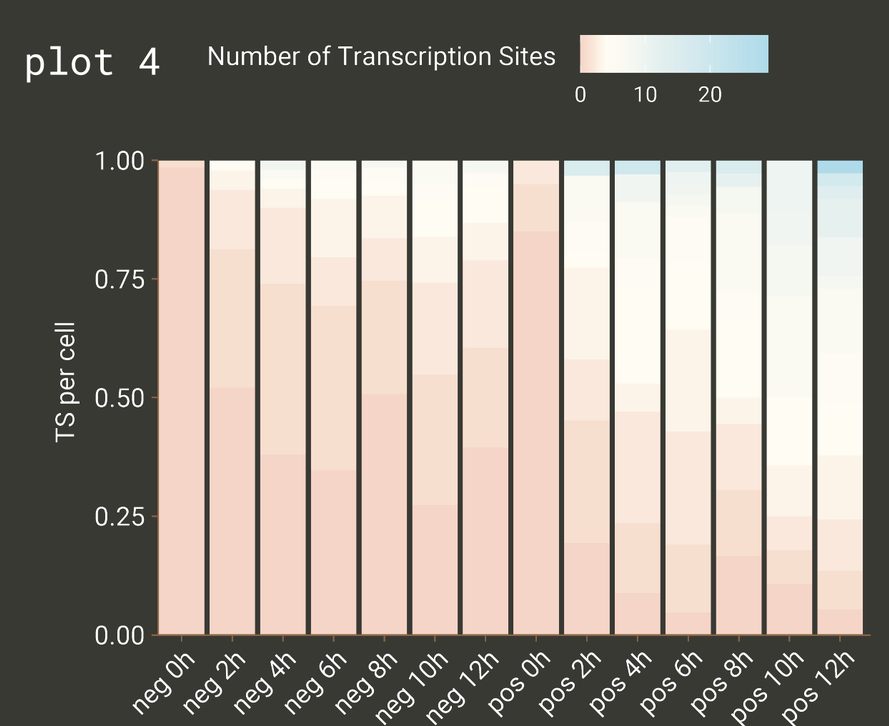
Number of Transcription Sites
TS per cell

Number of GBP1 mRNAs per cell at different time points
plot 5

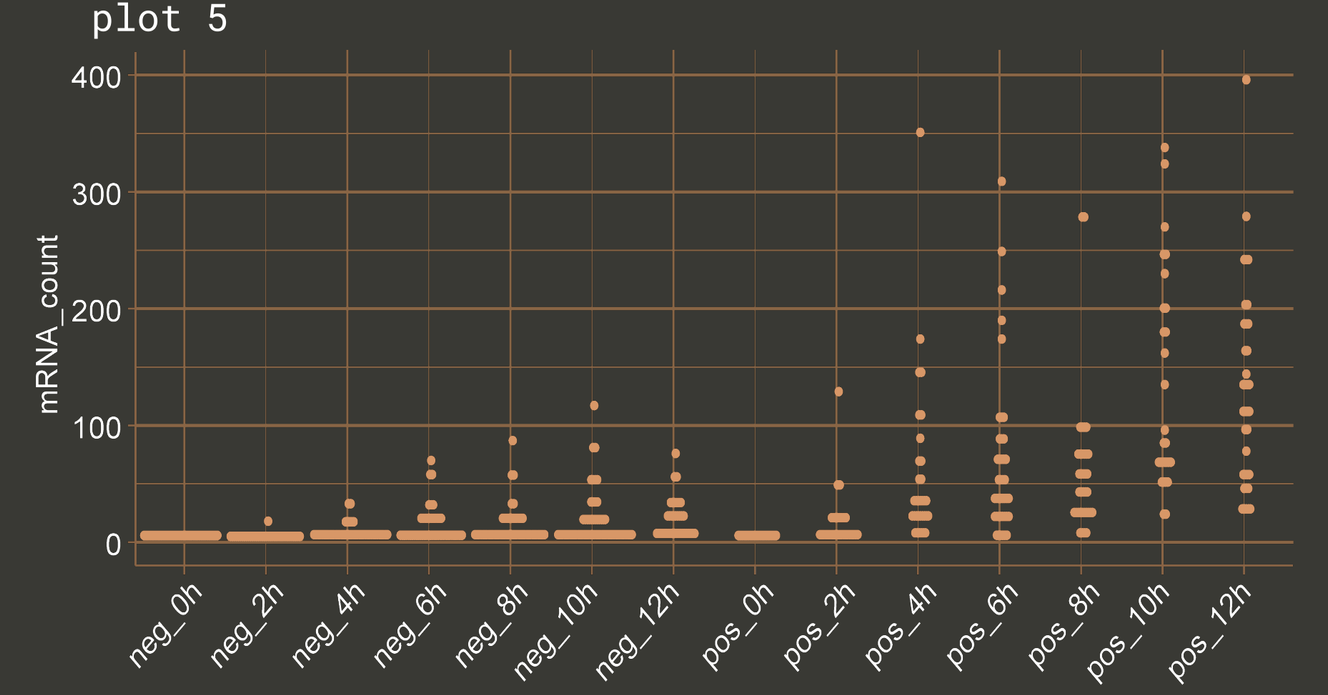
mRNA_count

plot 6
Distribution of TS Integrated densities for GBP1

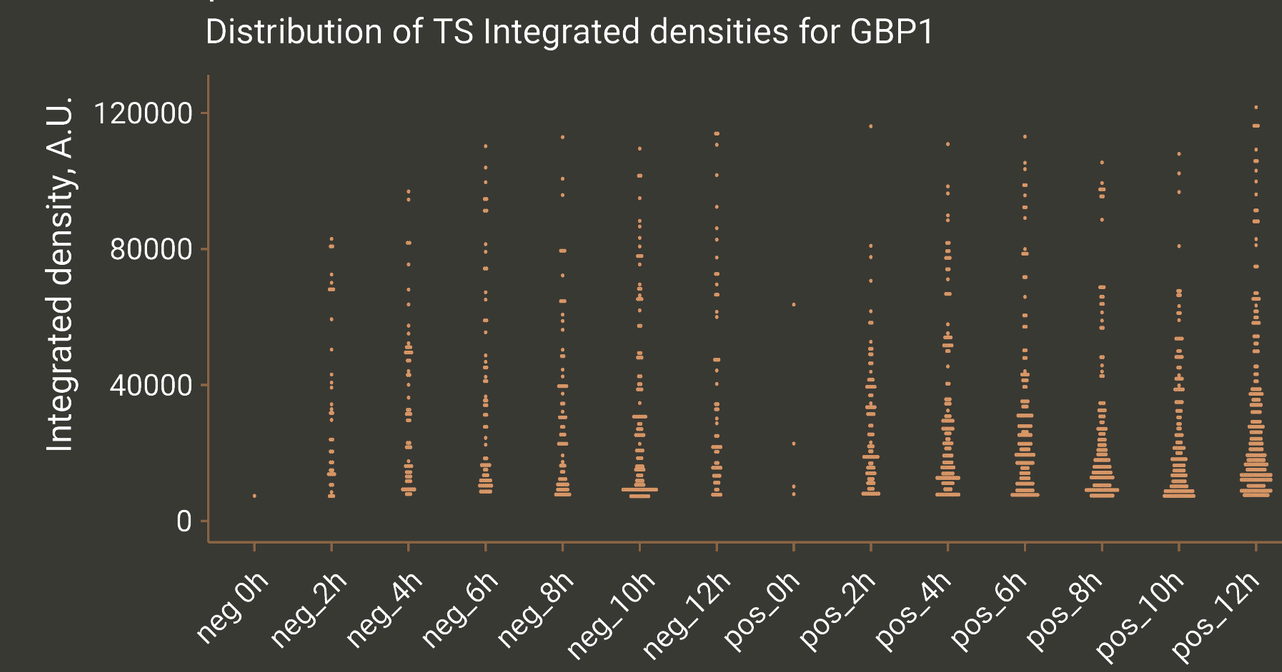
Integrated density, A.U.

GBP1 - transcription speed increased, unclear if transcription occurs earlier

GBP1 - скорость транскрипции увеличивается, но не понятно, начинается ли транскрипция раньше

Methods 2/
/Методы 2
GFP-reporter for GBP1 protein with a P2A sequence

Software used/
/Использованное программное обеспечение
FIJI
TrackMate-Cellpose
RStudio

Answer 1 - Transcription of GBP1 and GBP5 in primed cells both begins earlier and goes with increased speed

Ответ 1 - Транскрипция в клетках, которые обработаны интерфероном повторно, начинается раньше и идёт быстрее

experiment 2 - data for gene GBP1 - responce to INFγ was recorded as a video file

1st induction of interferon:
Первое внесение интерферона:

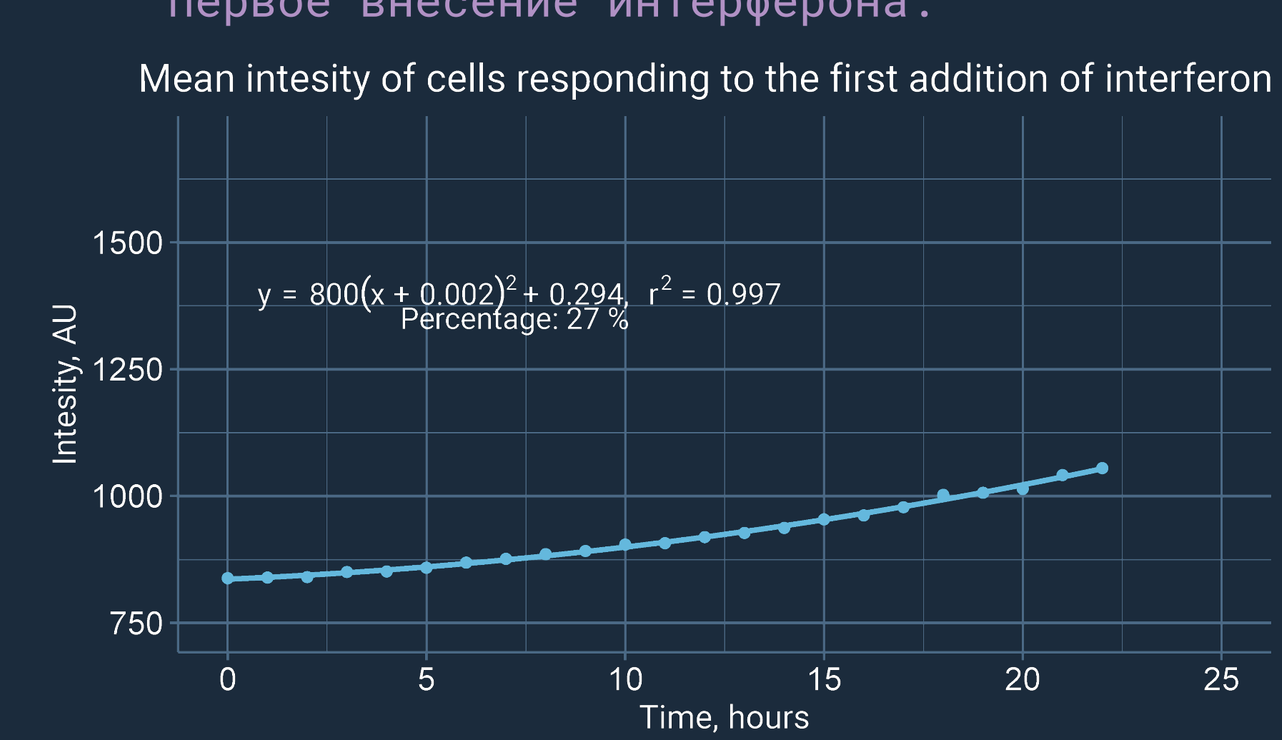
Mean intesity of cells responding to the first addition of interferon
y = 800(x + 0.002)² + 0.294, r² = 0.997
Percentage: 27 %
Intesity, AU
Time, hours

2nd induction of interferon:
Второе внесение интерферона:

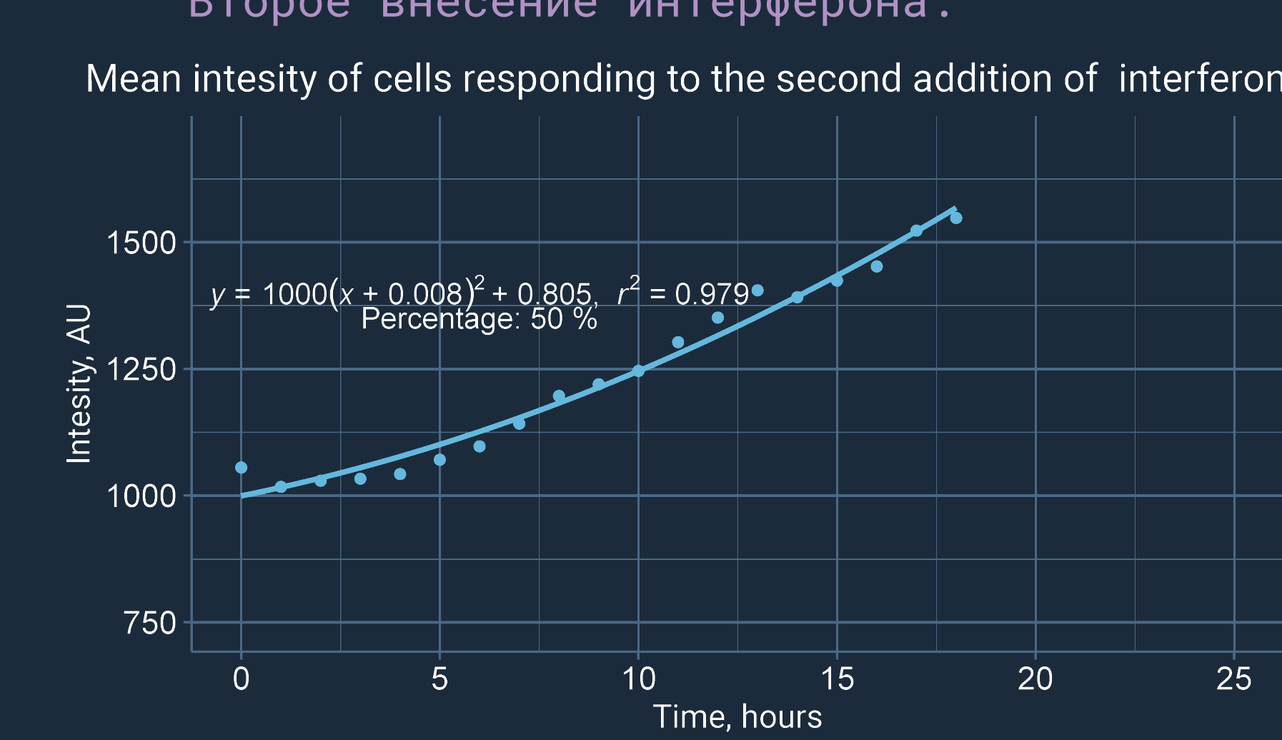
Mean intesity of cells responding to the second addition of interferon
y = 1000(x + 0.008)² + 0.805, r² = 0.979
Percentage: 50 %
Intesity, AU
Time, hours

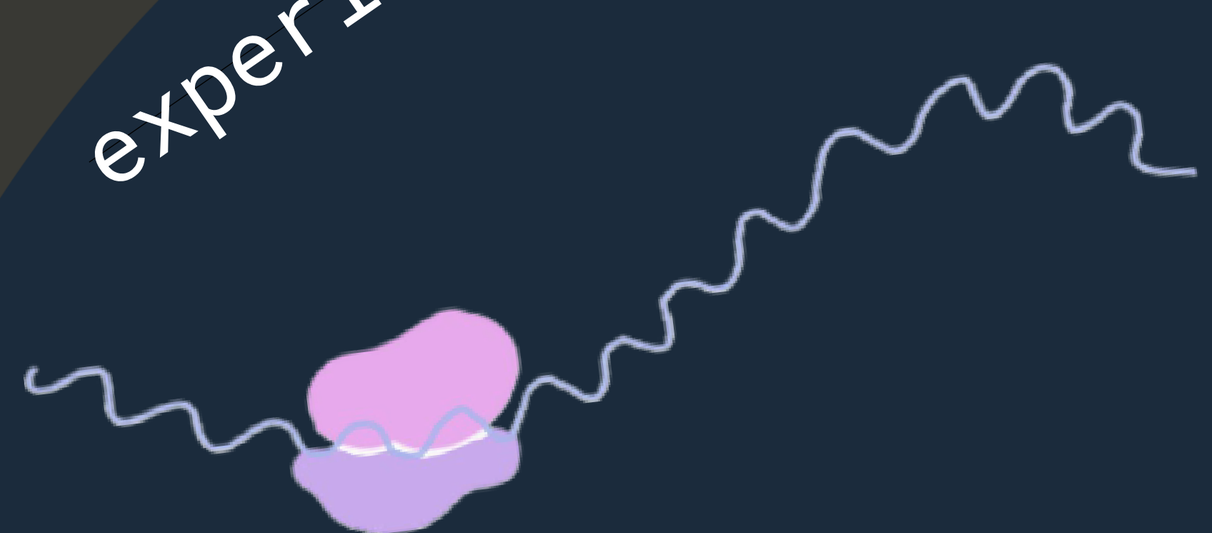

No interferon:
Без интерферона:

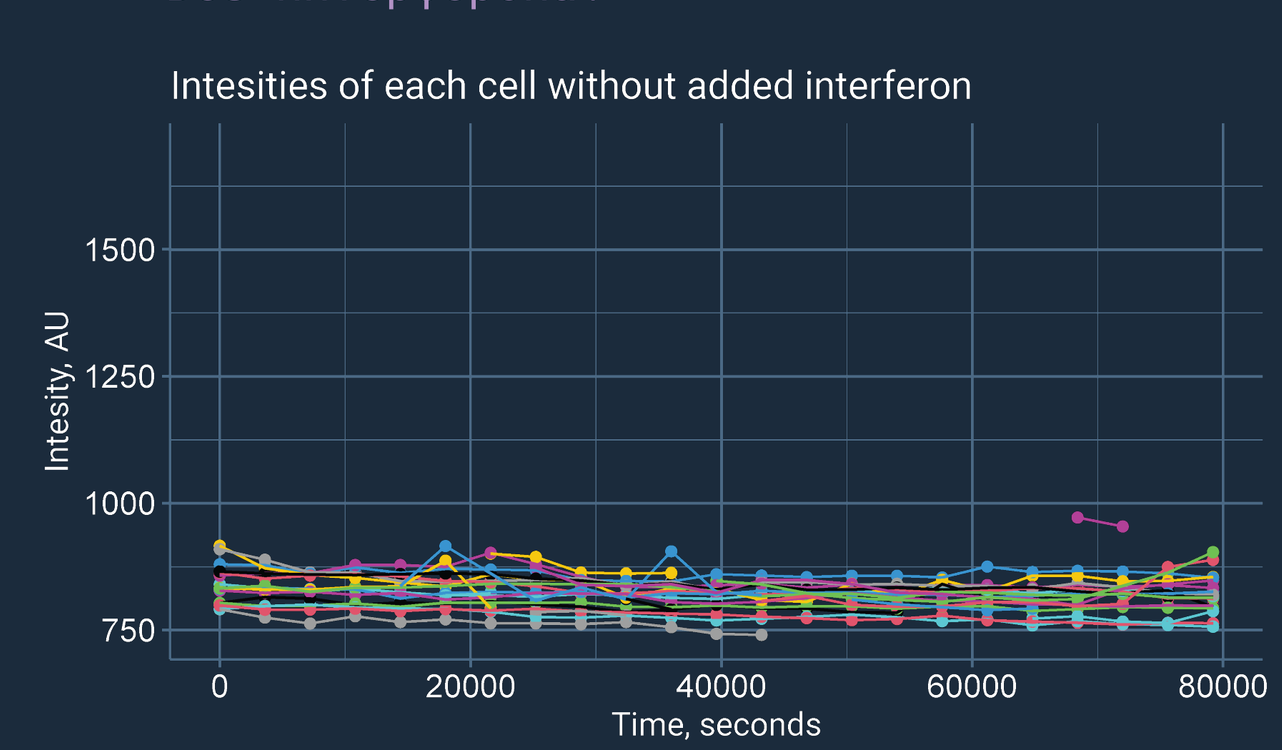
Intesities of each cell without added interferon
Intesity, AU
Time, seconds

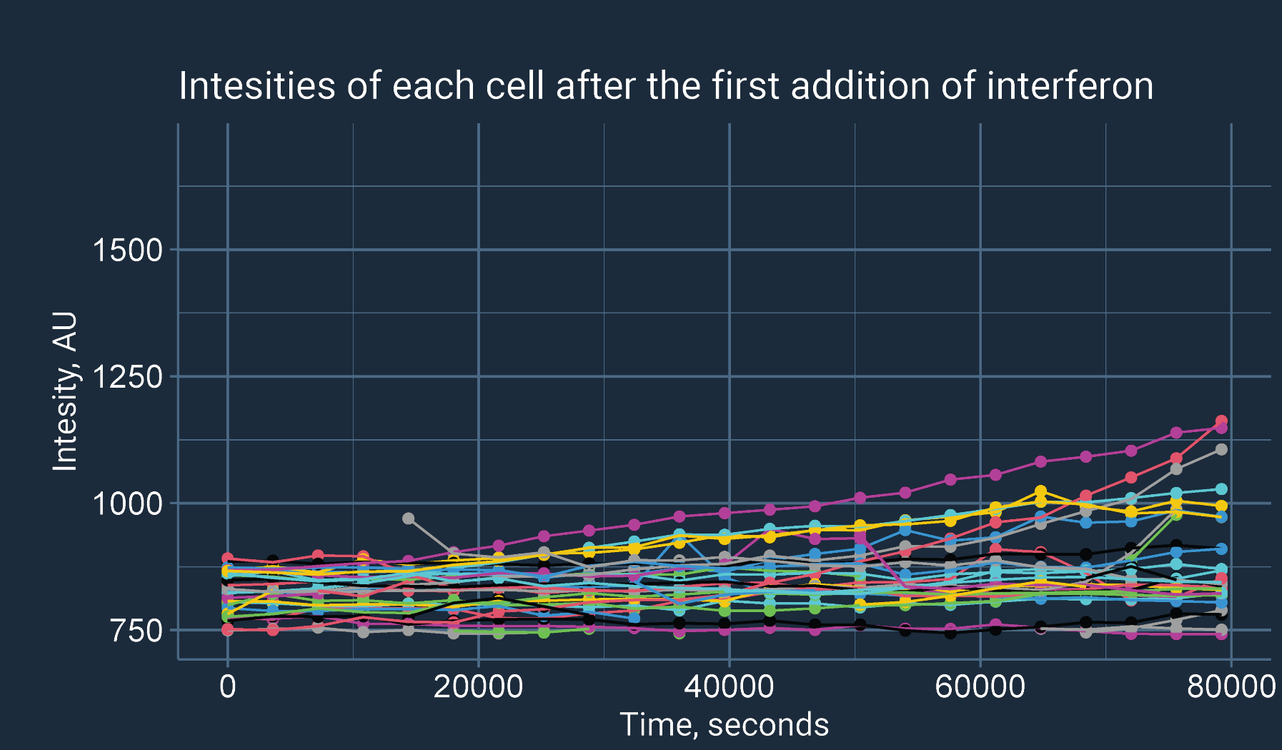
Intesities of each cell after the first addition of interferon
Intesity, AU
Time, seconds

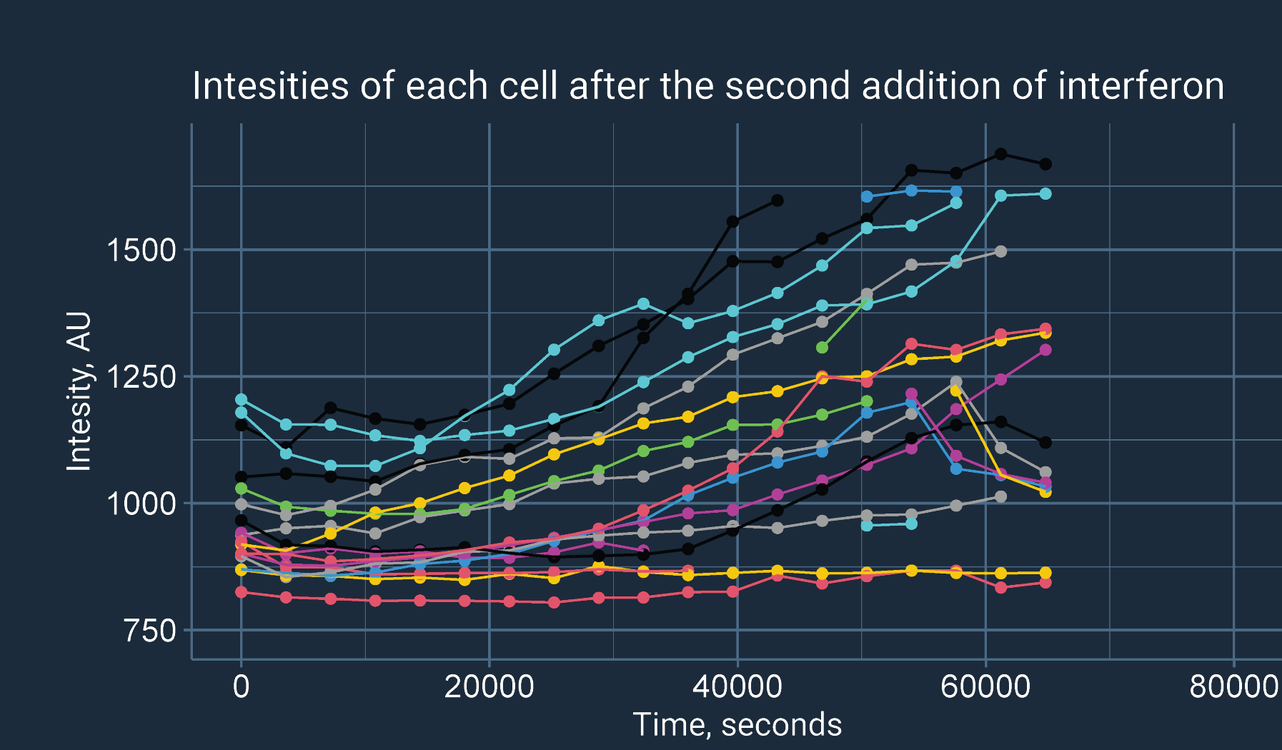
Intesities of each cell after the second addition of interferon
Intesity, AU
Time, seconds

Question 2.
Does translation begin earlier or go with increased speed? How does the percentage of expressing cells change?

Вопрос 2.
Трансляция белка начинается раньше или идёт с большей скоростью? Как изменился процент клеток, в которых идет трансляция?

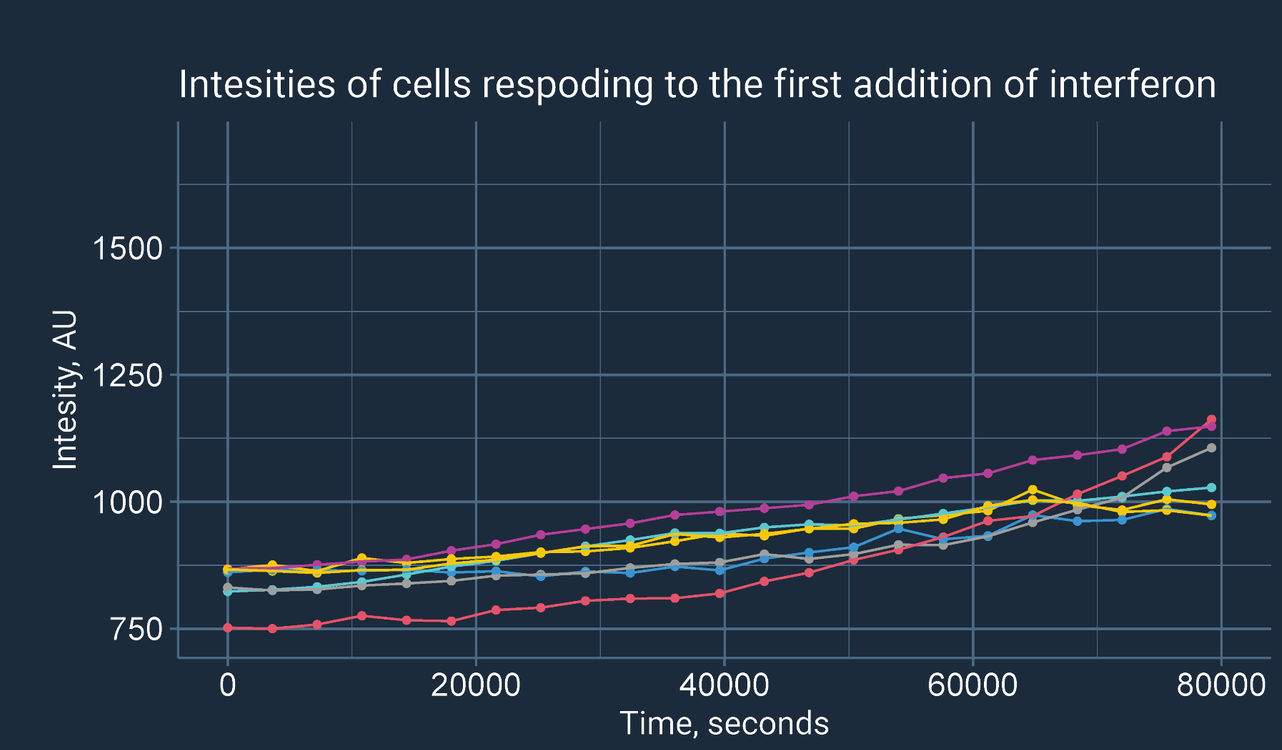
Intesities of cells respoding to the first addition of interferon
Intesity, AU
Time, seconds

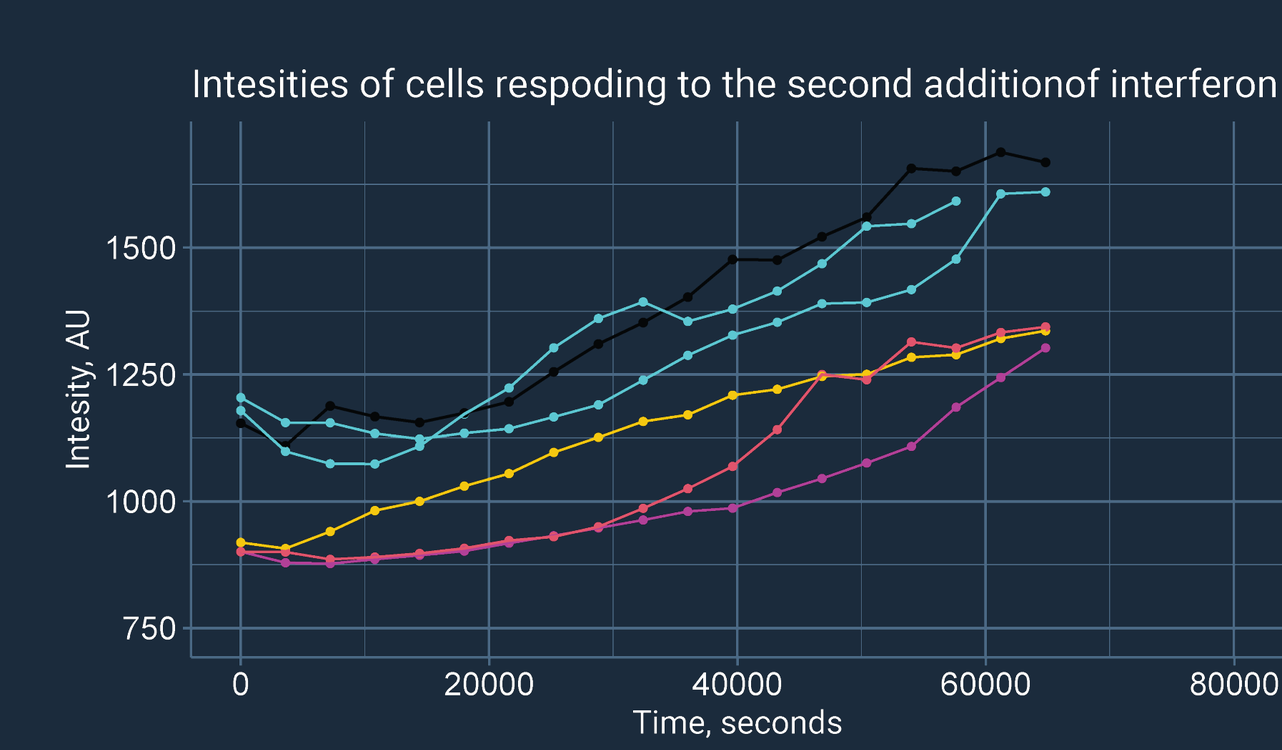
Intesities of cells respoding to the second additionof interferon
Intesity, AU
Time, seconds

Answer 2: We observed both an increase in translation speed and an earlier beginning of translation; the percentage of translating cells increased considerably. But! The cells could not fully recover from the first interferon induction, and the memory window should be further extended to eliminate this factor.

Ответ 2: Мы пронаблюдали как увеличение скорости трансляции так и более раннее её начало, и существенное увеличение транслирующих клеток; Но! было замечено что клетки не полностью востановились после первого индуцирования интерфероном, и необходимо увеличить временное окно чтобы исключить этот фактор.

conclusions and acknowledgments

We confirmed that transcriptional memory exists: upon second induction with interferon-gamma, the number of TS and intensity increases. The methods we used are now proven effective for studying transcriptional memory.
Мы подтвердили существование транскрипционной памяти — после повторного индуцирования интерфероном гамма количество сайтов транскрипции и яркость увеличиваются. Использованные нами методы оказались эффективны для изучения транскрипционной памяти.

software for video analysis
cell segmentation
analysis of microscopy data
→ intensity graphs + trendlines
equations describing translation rates

ПО для анализа видео
сегментация клеток
анализ данных
→ графики интенсивности
уравнения линии тренда

Further steps might include // Возможные дальнейшие шаги
Make trendline based on mean values of number of mRNA in cells // Сделать линию тренда по среднему числу мРНК в клетках
Get more time points to see if the signal reaches plato // Собрать больше данных, чтобы увидеть, достигает ли сигнал плато
Increase memory window between interferon inductions // Увеличить окно памяти между индуцированиями интерфероном.

Article about transcriptional memory
Статья о транскрипционной памяти

One of the main results of our project is development of R scripts for the analysis of this kind of data; in the future, they will be used on bigger datasets.
Один из главных результатов проекта — это разработка программ для анализа такого типа данных, в будущем, они будут использованы на более больших объемах данных

Acknowledgments//Благодарности
School of Molecular and Theoretical Biology
Zimin Foundation
HHMI - Howard Hughes Medical Institute
Tartu University

our sponsors

ZIMIN FOUNDATION
hhmi
SMTB
TARTU ÜLIKOOL · UNIVERSITAS TARTUENSIS · 1632