SMR 2024 Abstract P-091

First-in-Human Radiopharmaceutical Therapy (RPT) of [$^{203/212}$Pb]VMT01 for Patients with Pretreated Unresectable or Metastatic Melanoma (NCT05655312)

Morris ZS[1], Menda Y[2], Block MS[3], Wahl R[4], Osman MM[5], Arays R[6], Olszanski AJ[7], Mehr SA[8], Puhlmann M[9], Hanna A[9], Johnson FL[9], and Cho SY[1]

[1]University of Wisconsin, [2]University of Iowa, [3]Mayo Clinic, [4]Washington University, [5]St Louis University, [6]University of Kentucky, [7]Fox Chase Cancer Center, [8]Nebraska Cancer Specialists, [9]Perspective Therapeutics Inc.

Background

- Immune checkpoint inhibitors (ICI) are effective in melanoma, but many patients still progress after PD-1 therapy; PFS for current 2L+ therapies, including lifileucel, remains limited between 2.1 – 4.1 months[4,5,6,9]
- Melanocortin-1 receptor (MC1R) is a novel target for radiopharmaceutical therapy (RPT) and is found highly expressed on melanoma tissue [2]
- ^{203}Pb and ^{212}Pb is a promising theranostic isotope pair for RPT of cancer
- VMT01 is a MC1R-targeted RPT that can be radiolabeled with either ^{203}Pb (patient selection and dosimetry assessments) or ^{212}Pb (alpha particle therapy)
- In preclinical experiments [^{212}Pb]VMT01 demonstrated direct monotherapy activity with direct cell killing (cytoreduction); in addition, through induction of immunologic cell death (ICD) which is immunostimulatory, cooperation of [^{212}Pb]VMT01 and ICIs could be demonstrated[3]
- Here we present the initial safety, dosimetry and efficacy findings from the first two cohorts

Proposed Mechanism of Action for [^{212}Pb]VMT01 [7]

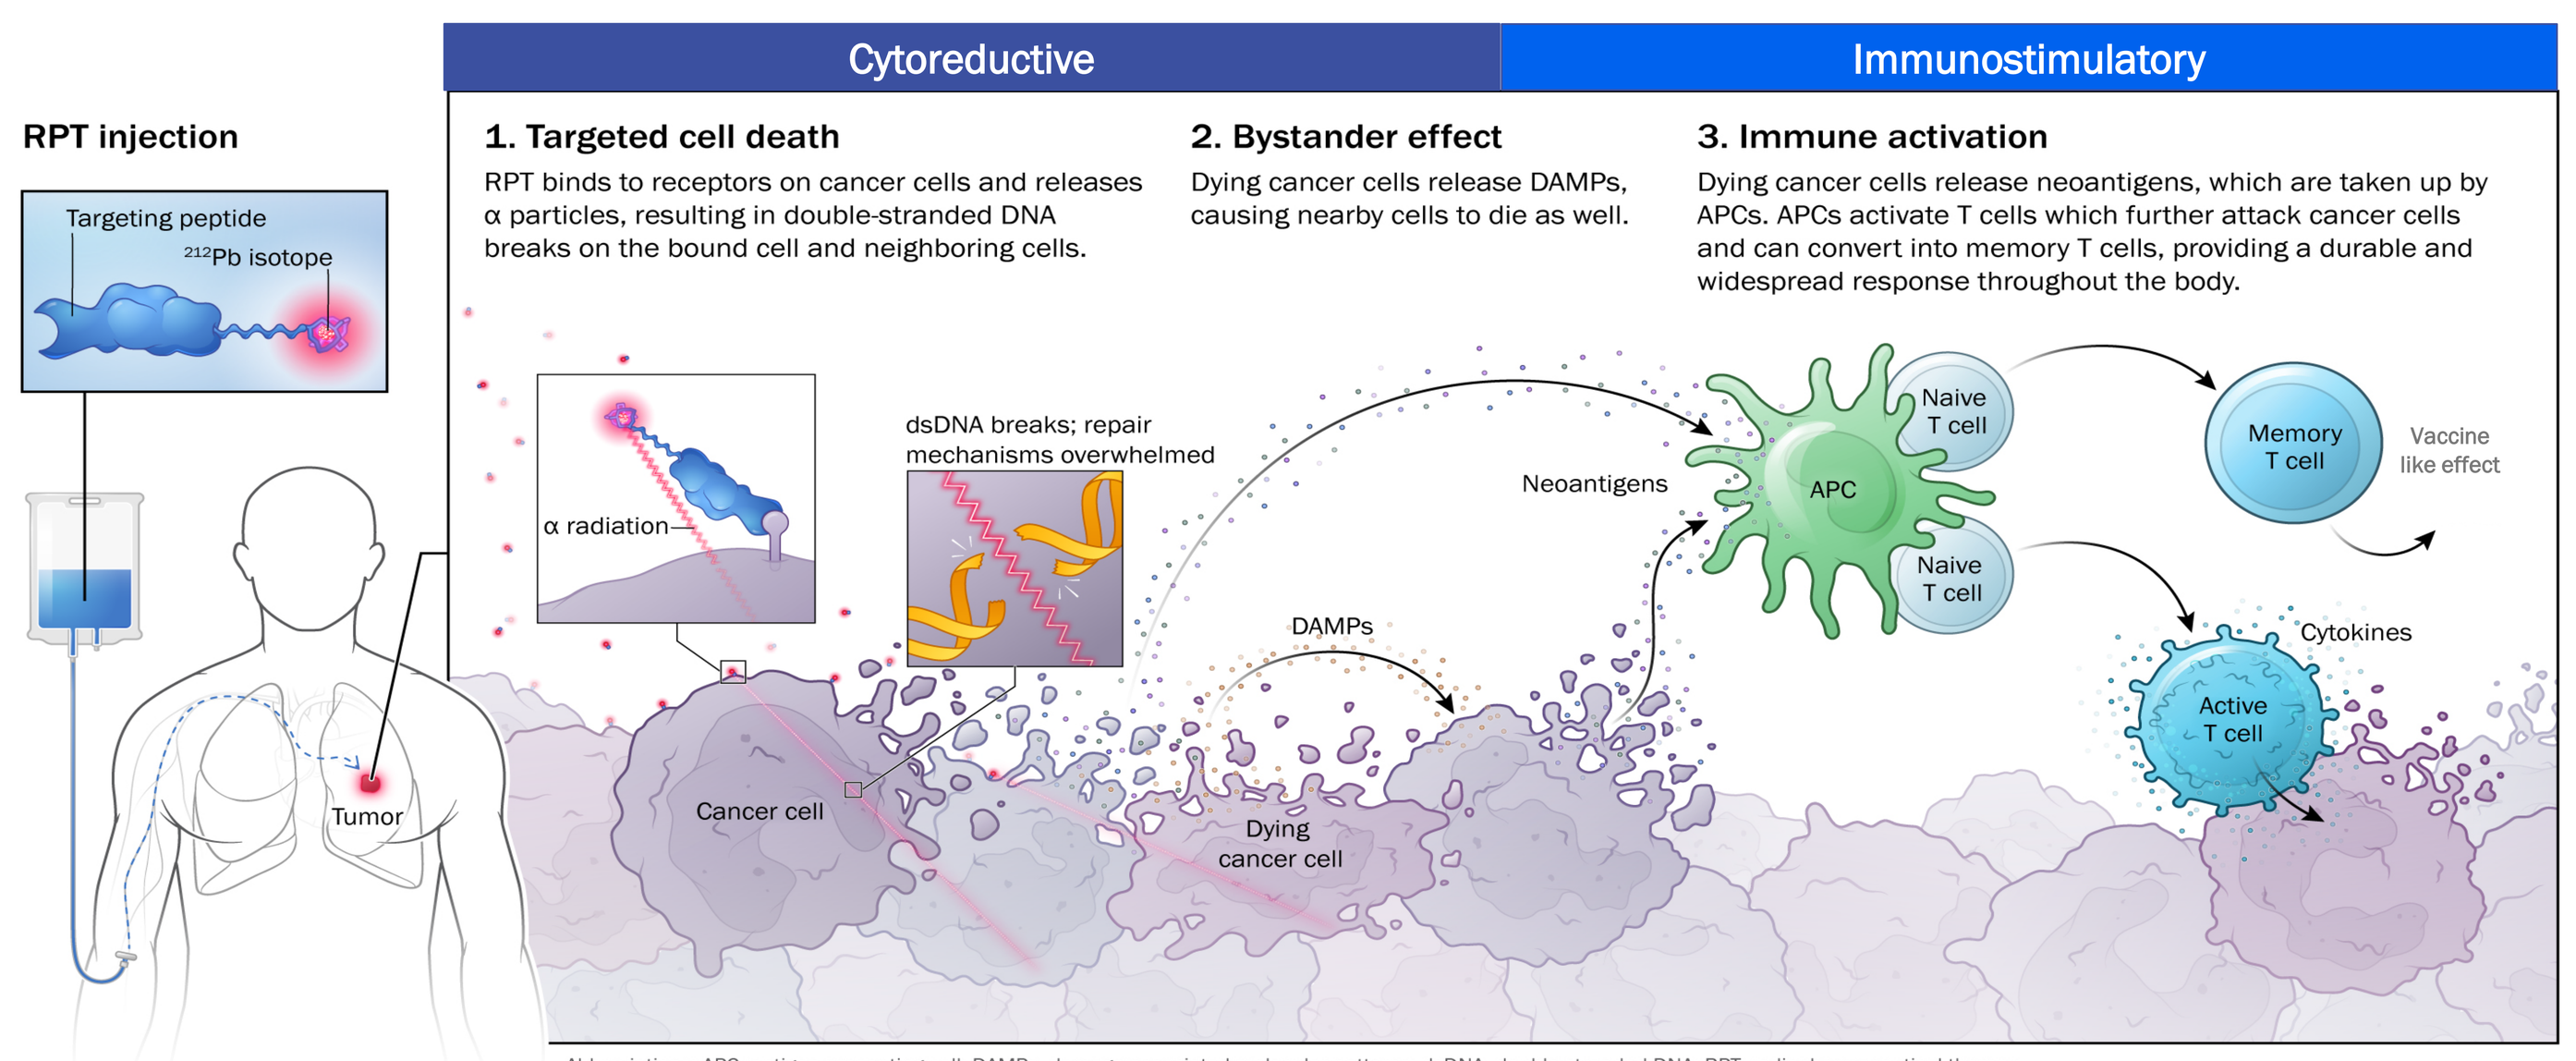

RPT injection
Targeting peptide
212Pb isotope
Tumor
Cytoreductive
Immunostimulatory
1. Targeted cell death
RPT binds to receptors on cancer cells and releases α particles, resulting in double-stranded DNA breaks on the bound cell and neighboring cells.
2. Bystander effect
Dying cancer cells release DAMPs, causing nearby cells to die as well.
3. Immune activation
Dying cancer cells release neoantigens, which are taken up by APCs. APCs activate T cells which further attack cancer cells and can convert into memory T cells, providing a durable and widespread response throughout the body.
α radiation
dsDNA breaks; repair mechanisms overwhelmed
DAMPs
Neoantigens
APC
Naive T cell
Naive T cell
Memory T cell
Vaccine like effect
Cytokines
Active T cell
Cancer cell
Dying cancer cell

Abbreviations: APC, antigen-presenting cell; DAMPs, damage-associated molecular patterns; dsDNA, double-stranded DNA; RPT, radiopharmaceutical therapy.

Study Design

- Patients were being selected for MC1R expression by [^{203}Pb]VMT01 SPECT/CT
- The first two cohorts incorporated voluntary, multiple-time points dosimetry using [^{203}Pb]VMT01 SPECT/CT scans
- Up to 3 treatment cycles of [^{212}Pb]VMT01, 8 weeks apart

Primary Objectives	To determine the MTD, MFD, and RP2D as mono- and in combination with PD-1 inhibitor (nivolumab)	MC1R Dosimetry	[^{203}Pb]VMT01 SPECT/CT
MC1R Imaging Patient Selection	[^{203}Pb]VMT01 SPECT/CT [^{68}Ga]VMT02 PET/CT	Design Methodology	mTPI-2

Monotherapy Dose Escalation	Cohort 1A [^{212}Pb]VMT01 N = 3 3 mCi (completed)	Cohort 2A [^{212}Pb]VMT01 N = 7 5 mCi (completed)	Cohort 3A [^{212}Pb]VMT01 N = 3-8 10 mCi	Cohort 4A [^{212}Pb]VMT01 N = 3-8 15 mCi	Potential Recommended Phase 2 Dose → Monotherapy expansion cohorts [^{212}Pb]VMT01 Up to N=25 Potential RP2D	Phase 2 expansion [^{212}Pb]VMT01 Monotherapy or [^{212}Pb]VMT01 + Nivolumab Combination Therapy Up to N=100 at RP2D
Combination Dose Escalation		Cohort 1B [^{212}Pb]VMT01 + Nivolumab N = 2-8 3 mCi or SMC Recommended dose	Cohort 2B [^{212}Pb]VMT01 + Nivolumab N = 2-8 5 mCi or SMC Recommended dose	Cohort 3B [^{212}Pb]VMT01 + Nivolumab N = 3-8 7.5 mCi or SMC Recommended dose	Combination expansion cohorts [^{212}Pb]VMT01 + Nivolumab Up to N=25 Potential RP2D	

Nivolumab will be administered 480 mg Q4W up to 24 months

Patient Characteristics (N=10)

Age	
N	10
Mean (SD)	67.2
Median	67
Range	49, 81
Sex, N (%)	
Female	4 (40)
Male	6 (60)
Race, N (%)	
White	10 (100)
Fitzpatrick Skin Phototype, N (%)	
TYPE I	2 (20)
TYPE II	1 (10)
TYPE III	2 (20)
TYPE IV	5 (50)
Time from diagnosis to C1D1 (yrs)	
N	10
Mean (SD)	5.1
Median	4.5
Min, Max	1.3, 10.4
ECOG Performance Status, N (%)	
0	6 (60)
1	4 (40)
Best Response to last systemic therapy, N (%)	
PD	4 (40)
SD	6 (60)

	Prior Lines Systemic Therapy (median)	Prior Lines IO Therapy (median)
Cohort 1 (3mCi) (N = 3)	5	3
Cohort 2 (5 mCi) (N = 7)	5	3

Adverse Events Summary

- No dose-limiting toxicities (DLTs) were observed among any patients
- Treatment-related AEs were low frequency (in 30% of patients), and all were of low grade (all Gr 1 except for a single case of Gr 2 anemia)
- One SAE was reported, hemoptysis. This was attributed to the patient's underlying condition (metastatic melanoma) and not related to [^{212}Pb]VMT01
- **No AEs led to treatment discontinuation**

Treatment-emergent Adverse Events (TEAEs)

TEAE by Preferred Term and Grade (Frequency >1 and / or Grade >1

	[^{212}Pb]VMT01 3 mCi (N=3)			[^{212}Pb]VMT01 5 mCi (N=7)			Total (N=10)		
Grade	Grade 1	Grade 2	Grade 3	Grade 1	Grade 2	Grade 3	Grade 1	Grade 2	Grade 3
Total Number of TEAEs Reported for All patients	31	6	-	12	12	4	43	18	4
AEs by Preferred Term and Grade Reported per Patient *[no of pts with AE (% of pts treated per cohort)]*									
Grade 1 and Frequency > 1	-	-	-	-	-	-	-	-	-
Anaemia	3 (100)	-	-	-	1 (14)	-	3 (30)	1 (10)	-
Lymphocyte count decreased	2 (67)	1 (33)	-	1 (14)	1 (14)	1 (14)	3 (30)	2 (20)	1 (10)
Nausea	2 (67)	-	-	1 (14)	1 (14)	-	3 (30)	1 (10)	-
Hyponatraemia	2 (67)	-	-	-	-	-	2 (20)	-	-
Platelet count decreased	2 (67)	-	-	-	-	-	2 (20)	-	-
Abdominal pain	-	-	-	-	1 (14)	-	-	1 (10)	-
Back pain	-	-	-	-	1 (14)	-	-	1 (10)	-
Grade ≥2	-	-	-	-	-	-	-	-	-
Cough worsening, not coded	-	-	-	-	1 (14)	-	-	1 (10)	-
COVID-19	-	2 (67)	-	-	-	-	-	2 (20)	-
Dysphagia	-	-	-	-	1 (14)	-	-	1 (10)	-
Dyspnoea (incl 'Dyspnea worsening, not coded')	-	1 (33)	-	-	1 (14)	-	-	2 (20)	-
Fatigue (incl 'Fatigue worsening, not coded')	-	-	-	-	2 (28.6)	1 (14)	-	2 (20)	1 (10)
Haemoptysis	-	-	-	-	-	1 (14)	-	-	1 (10)
Hepatic vein thrombosis	-	-	-	-	1 (14)	-	-	1 (10)	-
Pneumonitis, not coded	-	-	-	-	1 (14)	1 (14)	-	1 (10)	1 (10)
Sinusitis	-	1 (33)	-	-	-	-	-	1 (10)	-
Urinary tract infection	-	1 (33)	-	-	-	-	-	1 (10)	-

[^{203}Pb]VMT01 Theranostic Dosimetry for [^{212}Pb]VMT01 (3 Subjects)

Dosimetry Summary

- Dosimetry with [^{203}Pb]VMT01 shows high kidney estimates for [^{212}Pb]VMT01
- Mean estimated cumulative dose to kidneys for Cohort 1 subjects (3 cycles at 3 mCi) was 25.8 Gy
- Cumulative dose estimates to kidneys for Cohort 2 subjects (2 cycles at 5 mCi) was between 25.6 Gy and 29.6 Gy
- **Despite high dosimetry estimates to the kidneys, no renal toxicities have been reported to date**

ROI	Dose Coefficients (Gy/mCi) and Cumulative Dose to Date (Gy)						Mean Coefficient, & Est'd Cumulative Dose	
	01-116		02-117		03-114			
	Coefficient (Gy/mCi)	Cumulative 2 x 5 mCi (Gy)	Coefficient (Gy/mCi)	Cumulative 1 x 5 mCi (GY)	Coefficient (Gy/mCi)	Cumulative 2 x 5 mCi (Gy)	Coefficient Mean ± SD (Gy/mCi)	Cumulative 3 x 3 mCi (Gy)
Liver	0.22	2.21	0.12	0.62	0.15	1.52	0.17 ± 0.05	1.49
Kidneys	2.96	29.6	3.07	15.4	2.56	25.6	2.87 ± 0.27	25.8
Spleen	0.23	2.27	0.34	1.72	0.29	2.85	0.29 ± 0.06	2.57
Marrow	0.11	1.08	0.08	0.41	0.08	0.82	0.09 ± 0.02	0.81
Tumors	0.25	2.53	0.14, 0.15	0.70 - 0.77	0.17, 0.26, 0.28	1.72 - 2.83	0.21 ± 0.06	1.26 - 2.55

Note: MC1R is expressed in normal renal parenchyma.[9] Tumor dosimetry does not account for partial volume effects

References

1. SEER: The Surveillance, Epidemiology, and End Results Program [Internet]. National Cancer Institute, DCCPS. 20002018. [cited 2022 Aug 7] Available from: SEER*Explorer Application (cancer.gov)
2. Su DG, et al. *JCO Precis Oncol.* 2024 Apr:8:e2300702
3. Li M et al. *Cancers* 2021, 13, 3676
4. Ascierto PA et al. *JCO* 41:2724-35, 2023
5. Arance A et al. *JCO* 41:75-85, 2022
6. Chesney J et al. *J Immunothera Cancer* 2022;10;e005755
7. Pouget J-P and Constanzo *J. Front. Med.* 2021;8:692436
8. Goldlinger SM et al. E.J.Ca. 162:222-33, 2022
9. Qiao Y et al. Sci. Rep. 2016;6:27589

Preliminary Efficacy

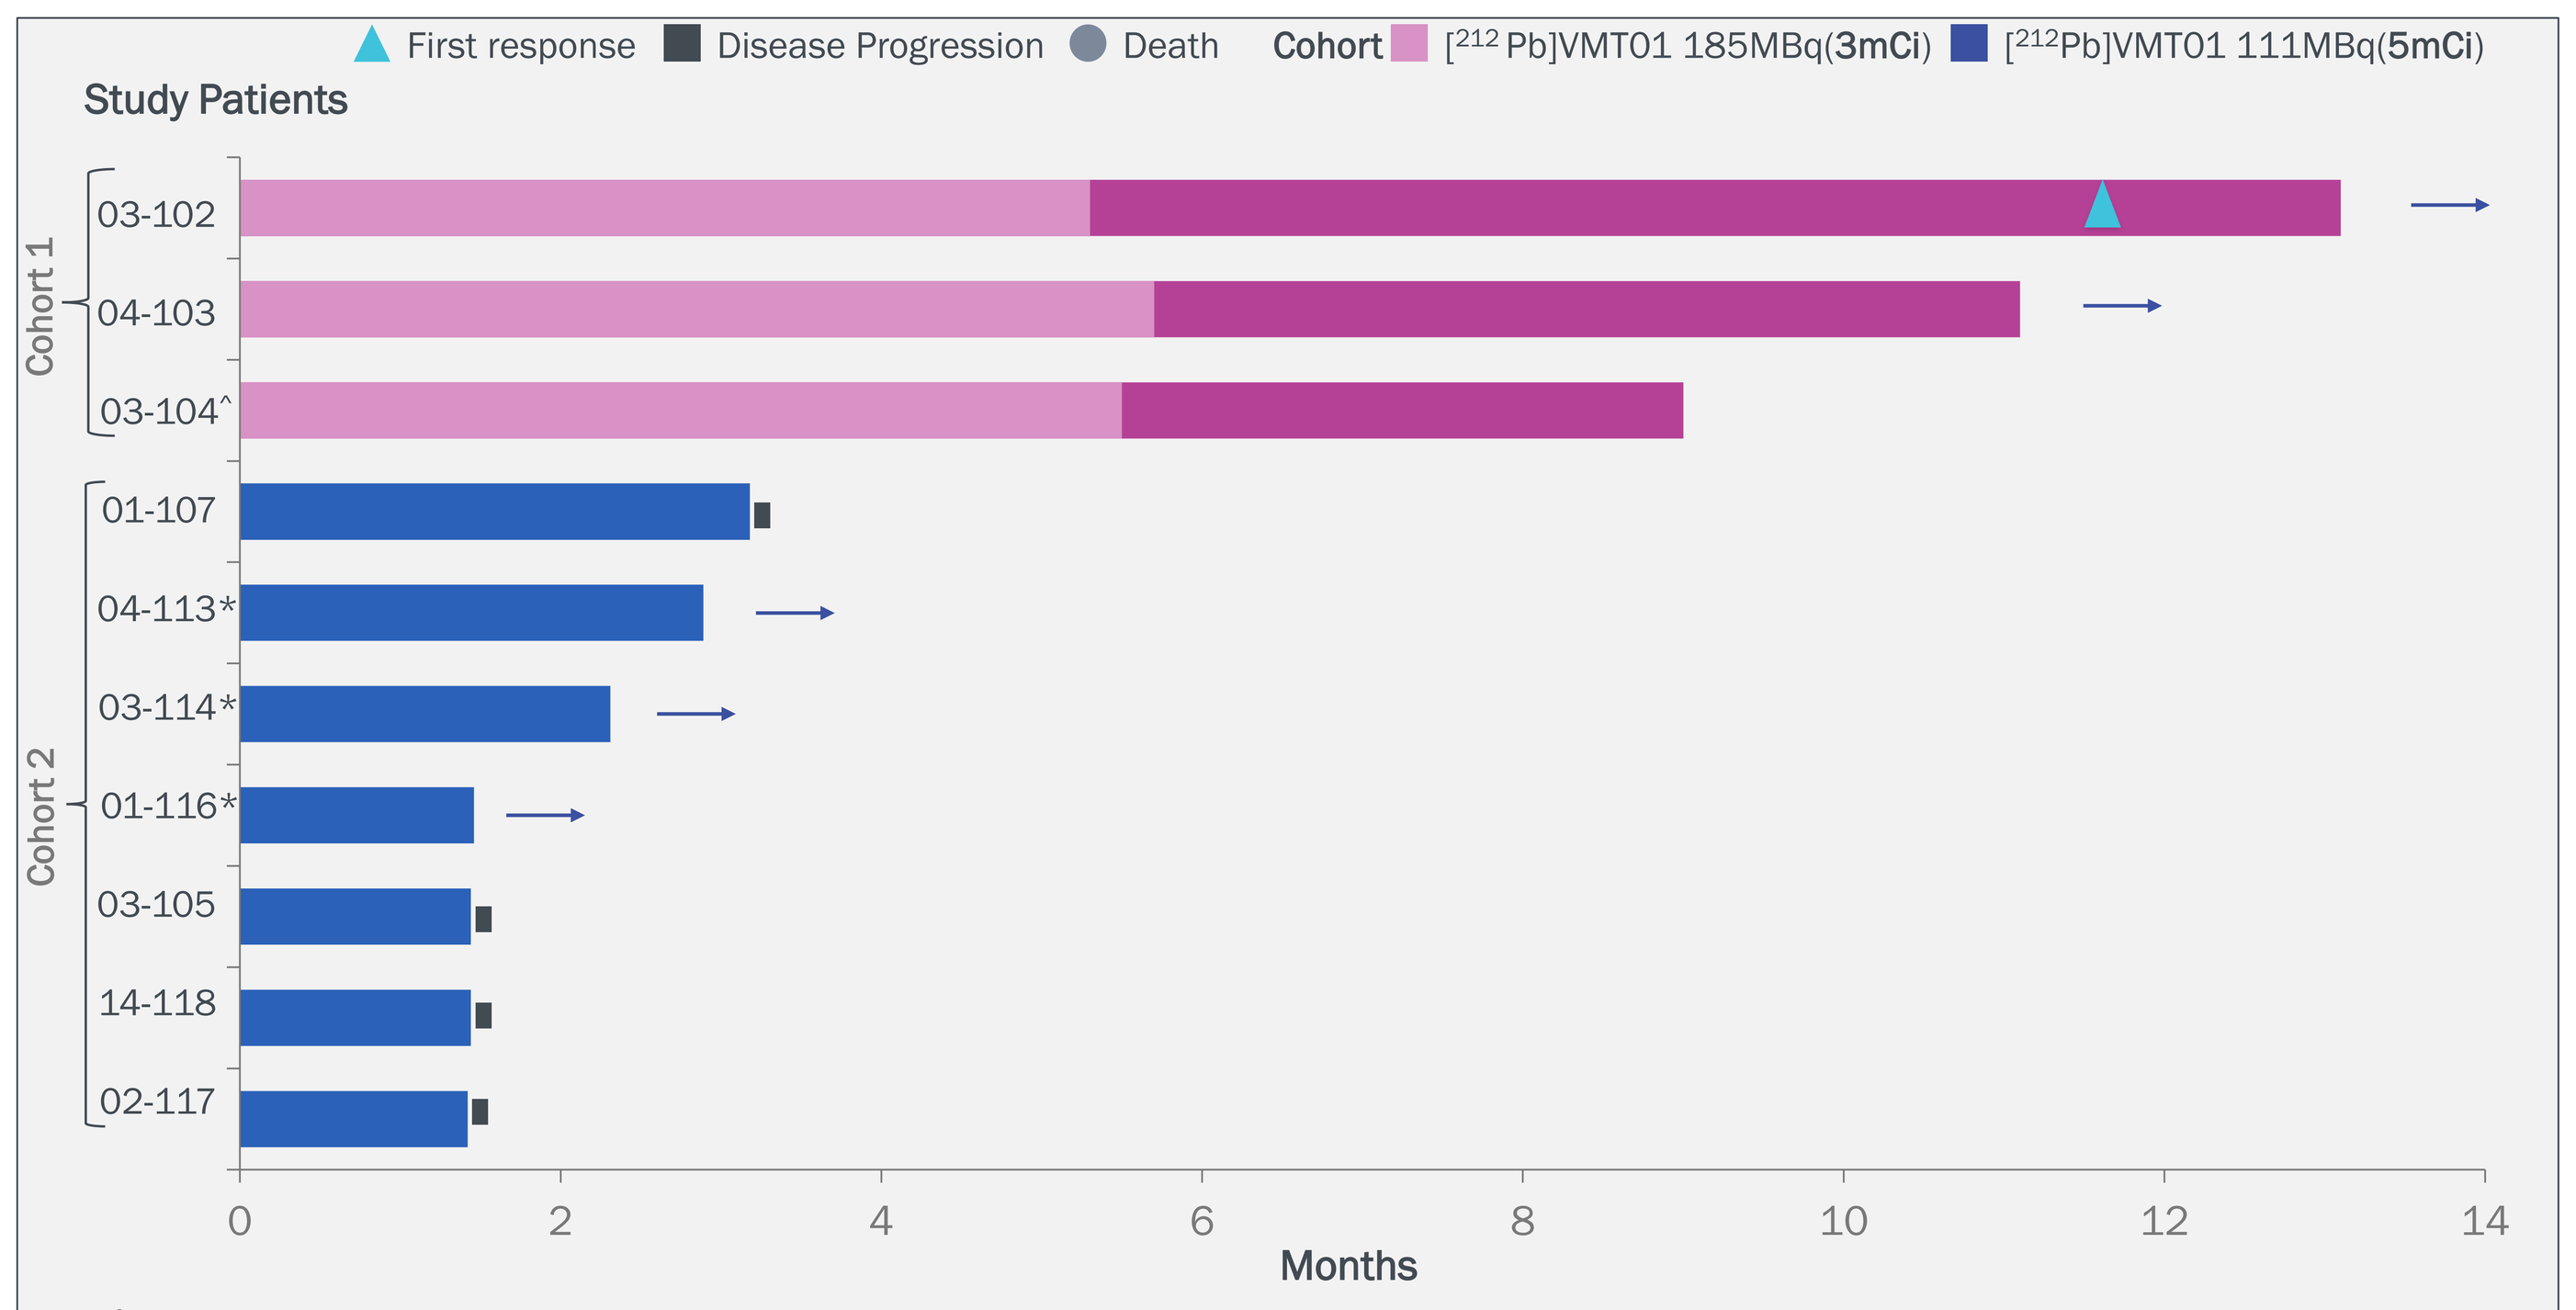

First response
Disease Progression
Death
Cohort
[212 Pb]VMT01 185MBq(3mCi)
[212Pb]VMT01 111MBq(5mCi)
Study Patients
Cohort 1
03-102
04-103
03-104^
Cohort 2
01-107
04-113*
03-114*
01-116*
03-105
14-118
02-117
0
2
4
6
8
10
12
14
Months

^Pt 03-104 discontinued imaging follow-up (without PD) to initiate a maintenance therapy
*Notes: Data cutoff (DCO) 04-SEP-2024. Patients 113, 114 and 116 had PD after DCO.

Patient with uPR after [^{212}Pb]VMT01

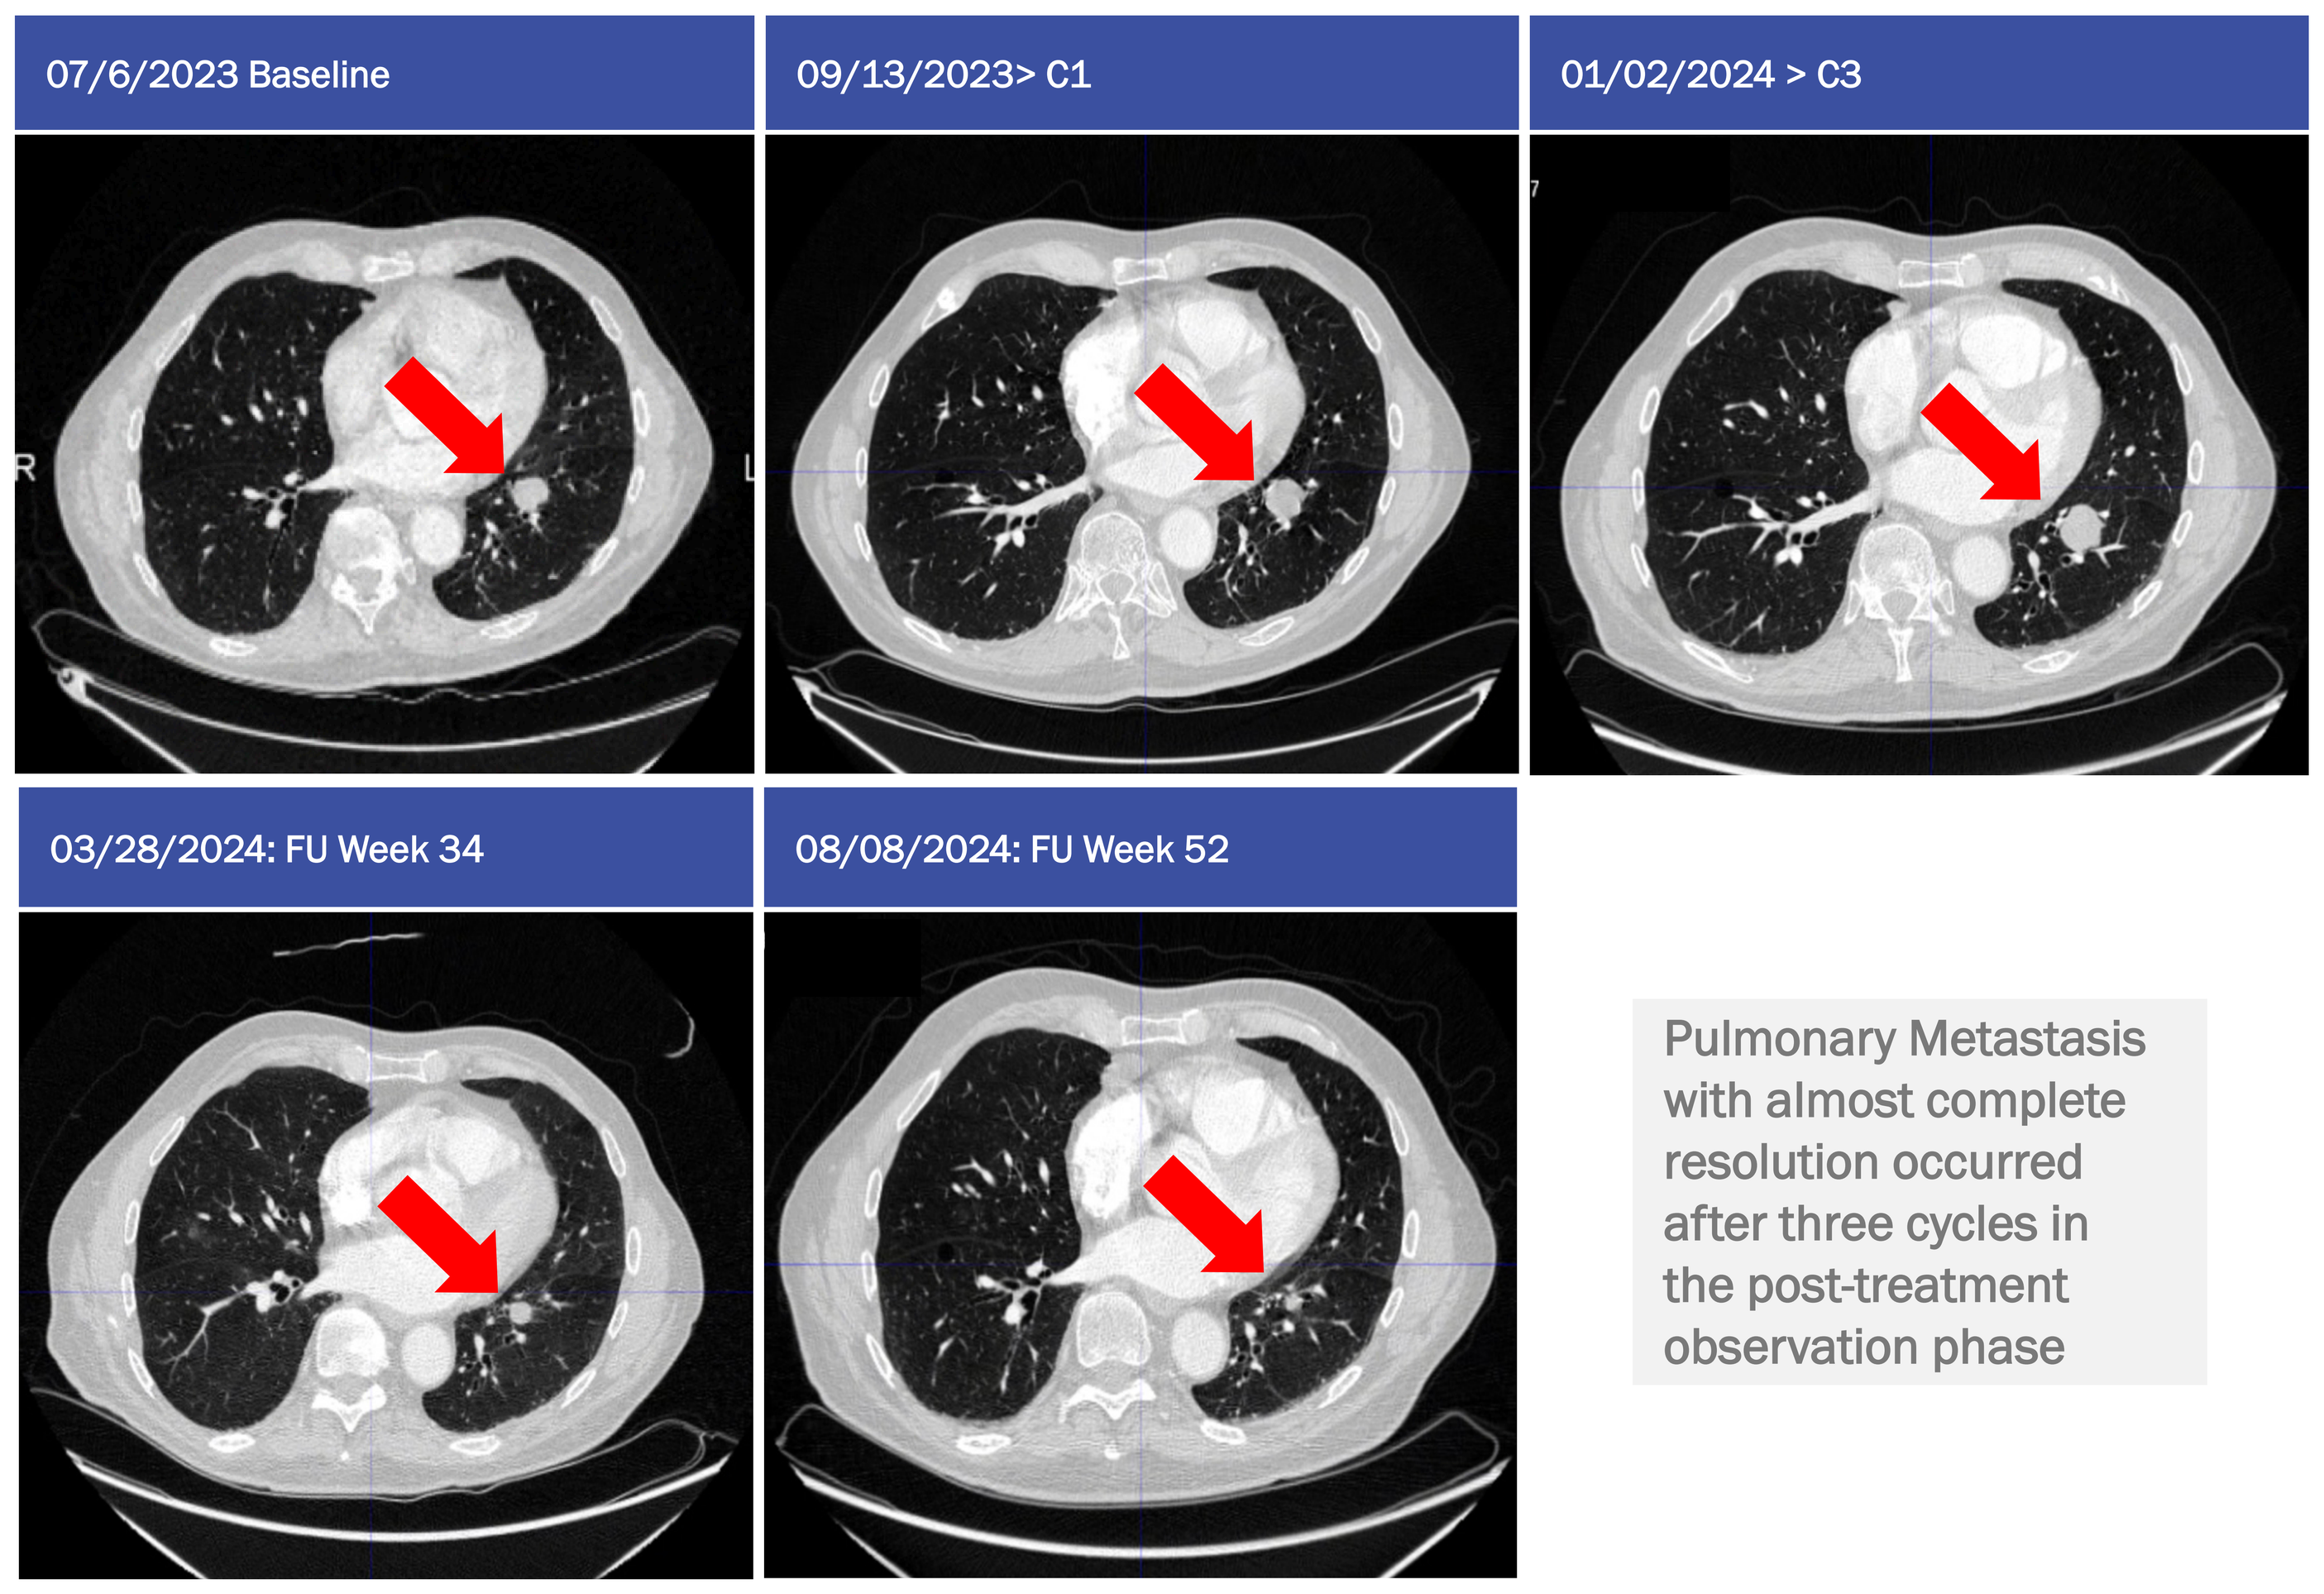

07/6/2023 Baseline
09/13/2023> C1
01/02/2024 > C3
03/28/2024: FU Week 34
08/08/2024: FU Week 52

Pulmonary Metastasis with almost complete resolution occurred after three cycles in the post-treatment observation phase

Efficacy Summary

- In Cohort 1, all subjects completed the planned treatment cycles; two subjects showed prolonged SD of 9 months and 11 months, respectively, from start of treatment
- One subject in Cohort 1 developed an objective response (PR – unconfirmed to date) after completion of all three VMT01 administrations and is still on trial after 13 months from start of treatment
- In Cohort 2, all subjects progressed after either the first cycle (3 subjects) or the second cycle (4 subjects)
- Preclinical experiments demonstrated higher immunostimulatory efficacy in combination with immune checkpoint inhibitors at lower doses whereas direct cytotoxic tumor effects were observed at higher activity doses[3]; here we see a delayed anti-tumor response at a lower dose with monotherapy
- We hypothesize that low-dose monotherapy in Cohort 1 leads to immunostimulatory activation within the tumor microenvironment (TME) resulting in prolonged disease control. This suggests the potential for the cooperation of [^{212}Pb]VMT01 with ICI in patients
- **Unexpectedly, we may have defined between Cohort 1 and Cohort 2 dose levels with VMT01 which may delineate the immunostimulatory and immunosuppressive effects on the TME in melanoma**

Conclusion

- At 3 mCi and 5 mCi activity levels, [^{212}Pb]VMT01 was safe and no DLTs were observed
- Dosimetry demonstrates high kidney estimates limiting monotherapy escalation
- SMC recommendation included dose reduction to 1.5 mCi for both monotherapy and the upcoming combination cohort with nivolumab
- **An objective response and prolonged PFS was observed at the 3 mCi activity level**
- The observed efficacy in Cohort 1 may be a result of the immunostimulatory effect of low dose ^{212}Pb-targeted alpha particle radiation on the TME, as observed in preclinical studies
- An amendment to test lower dose levels for monotherapy is planned
- **The combination cohort with nivolumab is active and now open for enrollment**

Acknowledgement

We would like to thank all participants, caregivers, investigators and site personnel. Dosimetry analysis was performed by Ian Marsh, and medical writing and graphical support was provided by Lucia Baratto, Stephen Keefe and Amos Hedt from Perspective Therapeutics.

This trial is sponsored by PERSPECTIVE THERAPEUTICS