Effectiveness of 12 or 24 week LDV-SOF and 12 week LDV-SOF + RBV in Treatment-Experienced Patients with Cirrhotic, Genotype 1 Hepatitis C: Real-world Experience from the TRIO Network

Michael P. Curry[1], Bruce Bacon[2], Douglas Dieterich[3], Steven L. Flamm[4], Lauren Guest[5], Kris V. Kowdley[6], Yoori Lee[5], Naoky Tsai[7], Zobair Younossi[8]

[1]Beth Israel Deaconess Medical Center, [2]Saint Louis University School of Medicine, [3]Mount Sinai School of Medicine, [4]Northwestern University Feinberg School of Medicine, [5]Trio Health Analytics, [6] Swedish Liver Center and Transplant Program, Swedish Medical Center, [7] Queens Medical Center, University of Hawaii, [8]Center for Liver Diseases, Department of Medicine, Inova Fairfax Hospital

Trio Health

1. BACKGROUND

The current recommendations for treatment of cirrhosis in HCV genotype 1 patients are dependent on prior treatment. 12 week ledipasvir-sofosbuvir (LDV-SOF) is recommended in treatment-naïve patients whereas 24 week LDV-SOF is recommended for prior treatment failures. In addition, 12 week LDV-SOF + RBV has been suggested as an alternative to 24 week LDV-SOF for treatment failures (Bourlière M, et al. Abs #82, AASLD 2014). No data is really available comparing these regimens in real-world patients that previously failed treatment.

2. AIM

To evaluate real-world SVR12 in treatment failure patients with HCV genotype 1 and cirrhosis treated with 12 or 24 weeks of LDV-SOF or 12 weeks of LDV-SOF + RBV.

3. METHODS

Data were obtained through Trio Health's Innervation Platform, a cloud-based disease management platform, and directly from specialty pharmacies including AcariaHealth, Allcare Plus, Aureus Health, BioCure, Encompass Rx, IslandCare, Pharmacare Hawaii, Premier, Skyemed, and other pharmacies for 476 treatment-experienced, genotype 1 patients with cirrhosis that initiated LDV-SOF ± RBV between Oct 2014 and Mar 2015. 45% (216/476) of the patients were treated in community practices with the remainder from academic centers.

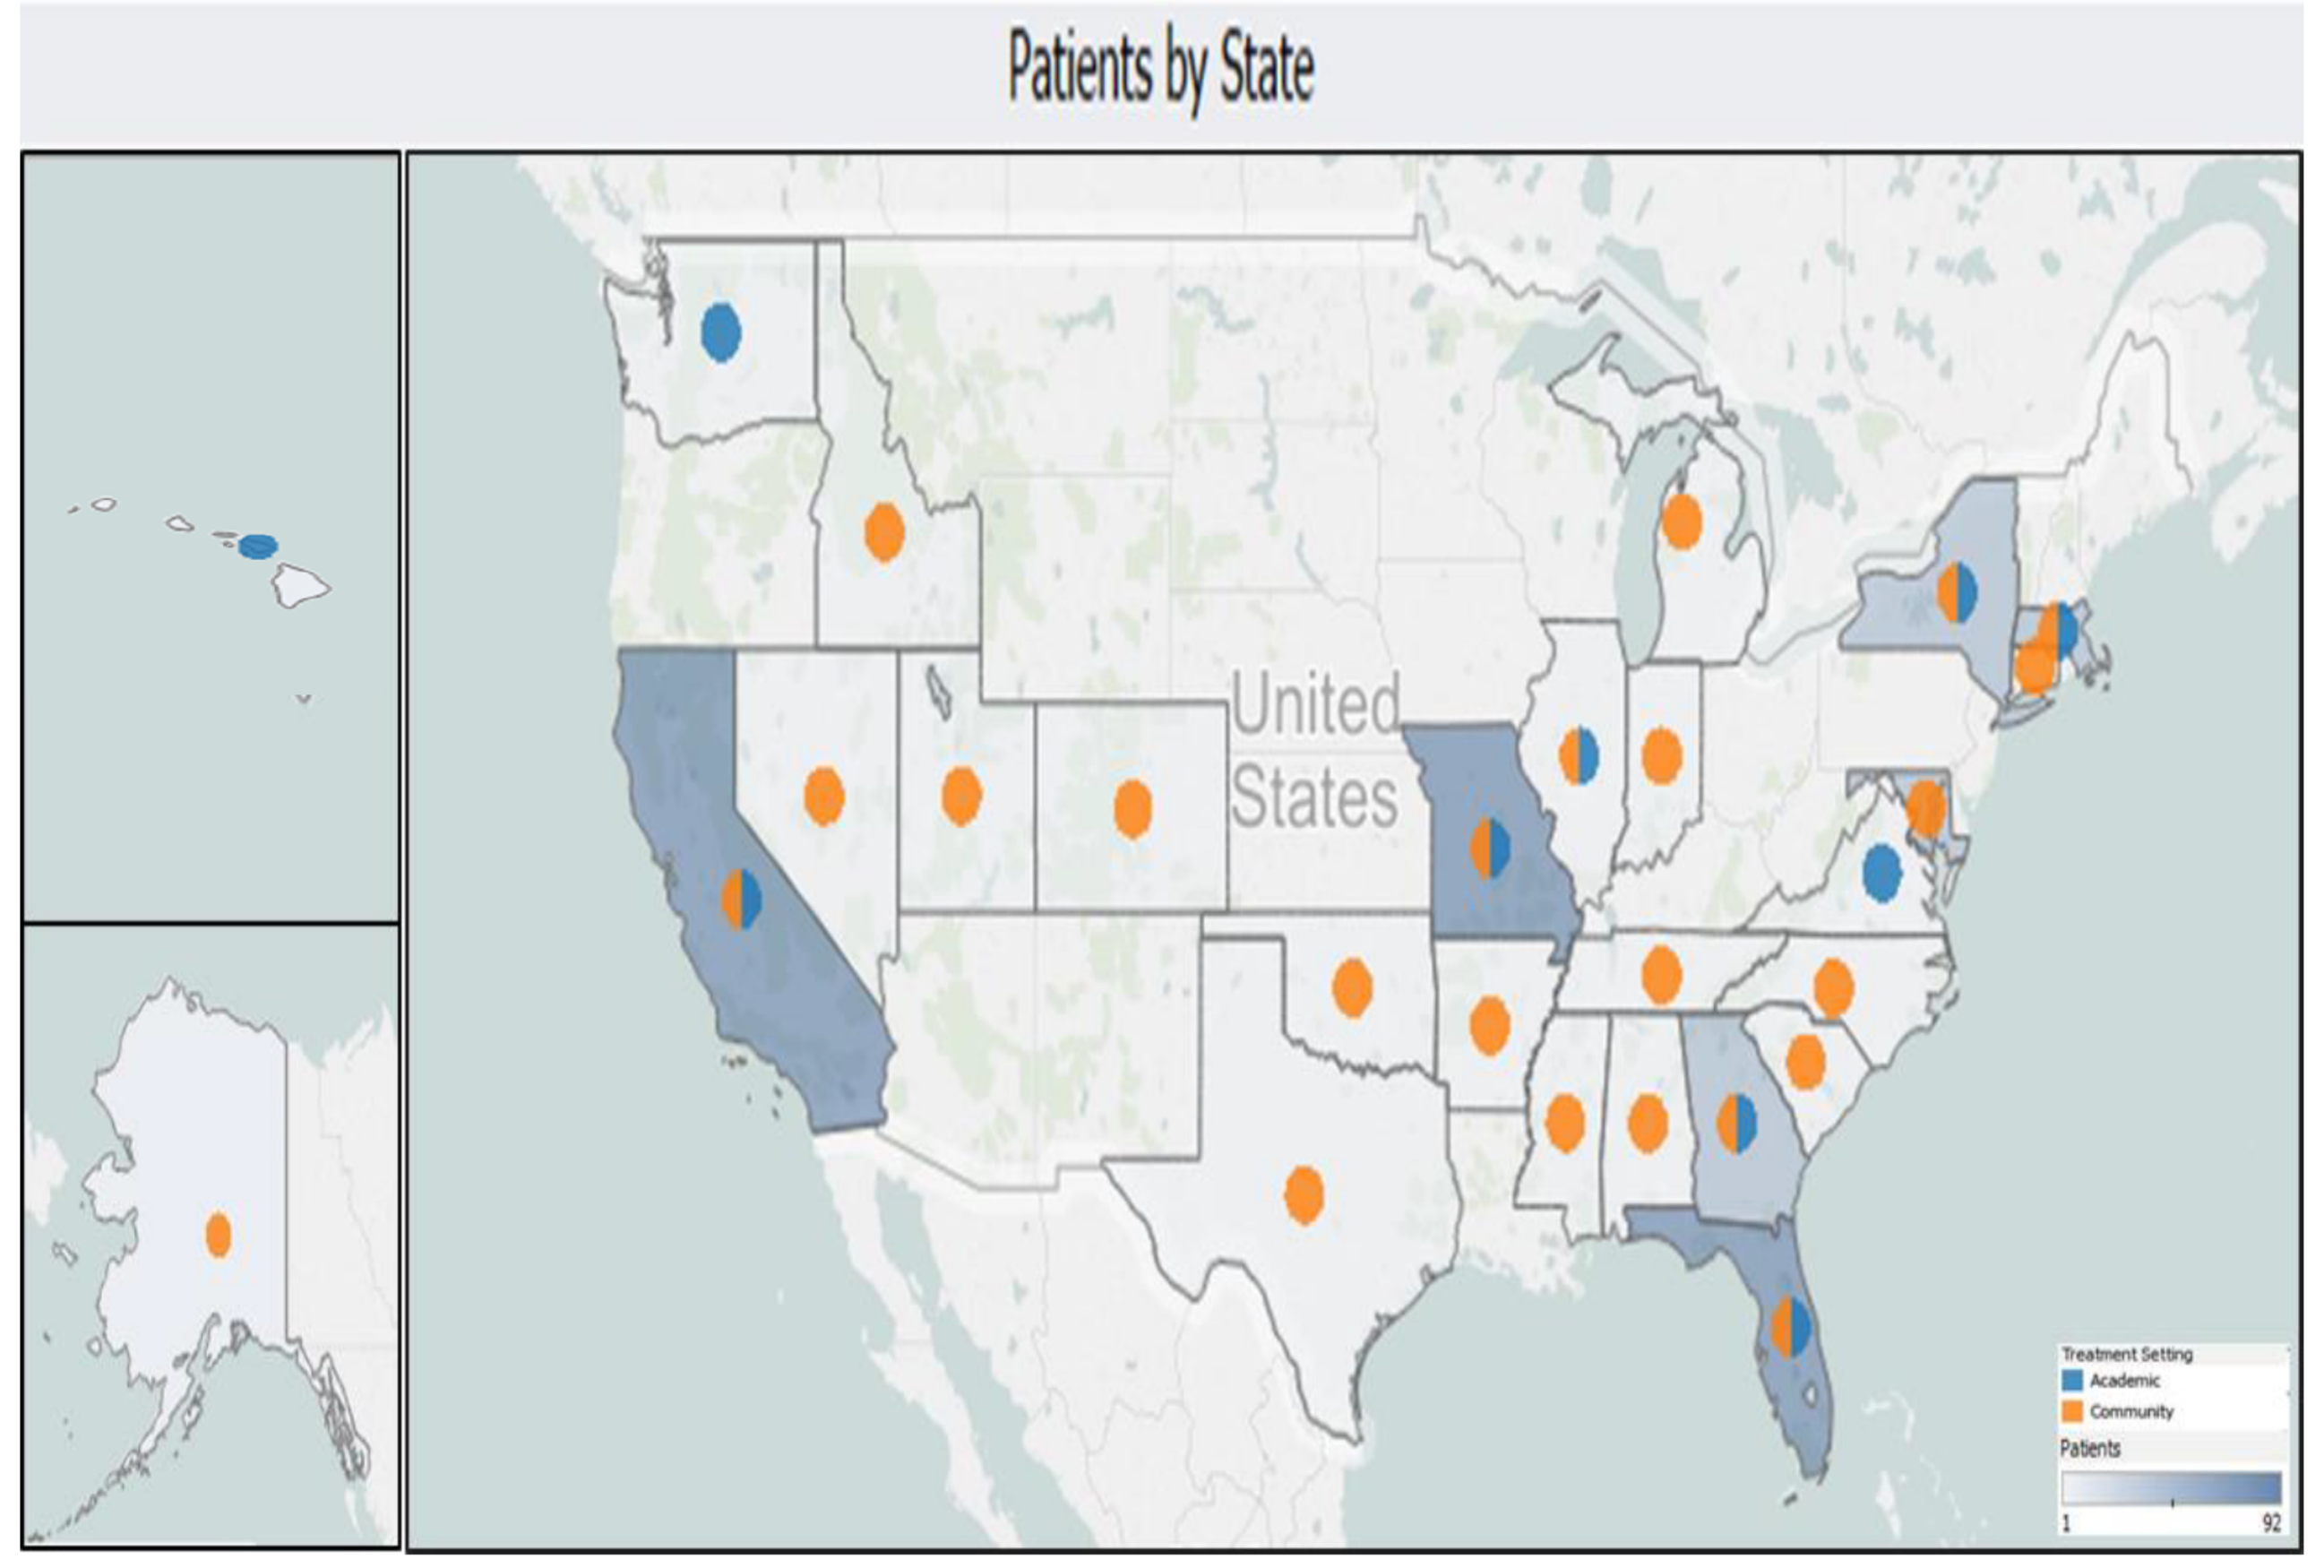
Patients by State
United States
Treatment Setting
Academic
Community
Patients
1
92

	LDV-SOF 12 week	LDV-SOF + RBV 12 Week	**LDV-SOF 24 week	Grand Total
Academic - no. (%)	50 (41%)	21 (81%)	189 (57%)	260 (55%)
Community - no. (%)	71 (59%)	5 (19%)	140 (43%)	216 (45%)
Total Count	121	26	329	476

**8 of 329 were treated with 24 weeks of LDV-SOF+RBV

12 Week Regimen by Practice Type

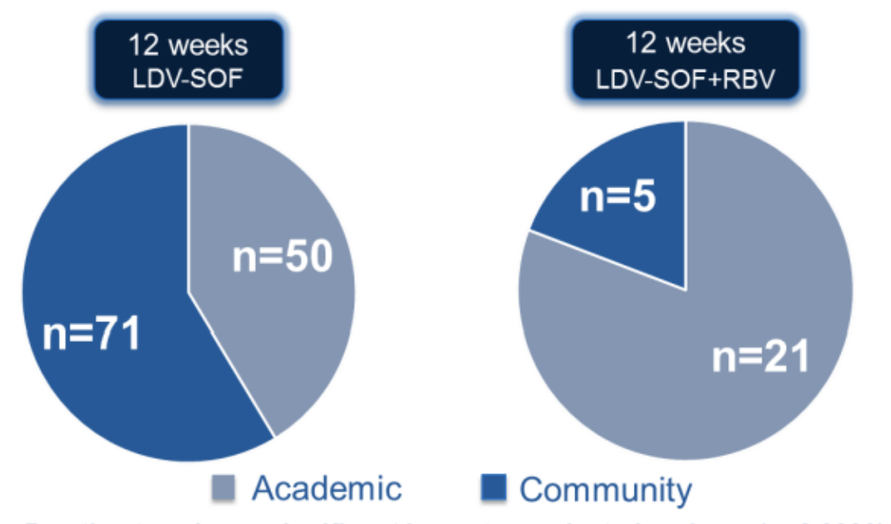
12 weeks LDV-SOF
12 weeks LDV-SOF+RBV
n=50
n=71
n=5
n=21
Academic
Community

Practice type has a significant impact on selected regimen (p=0.0003)
2-side asympt. p-value via Pearson χ^2.

4. PATIENT DISPOSITION

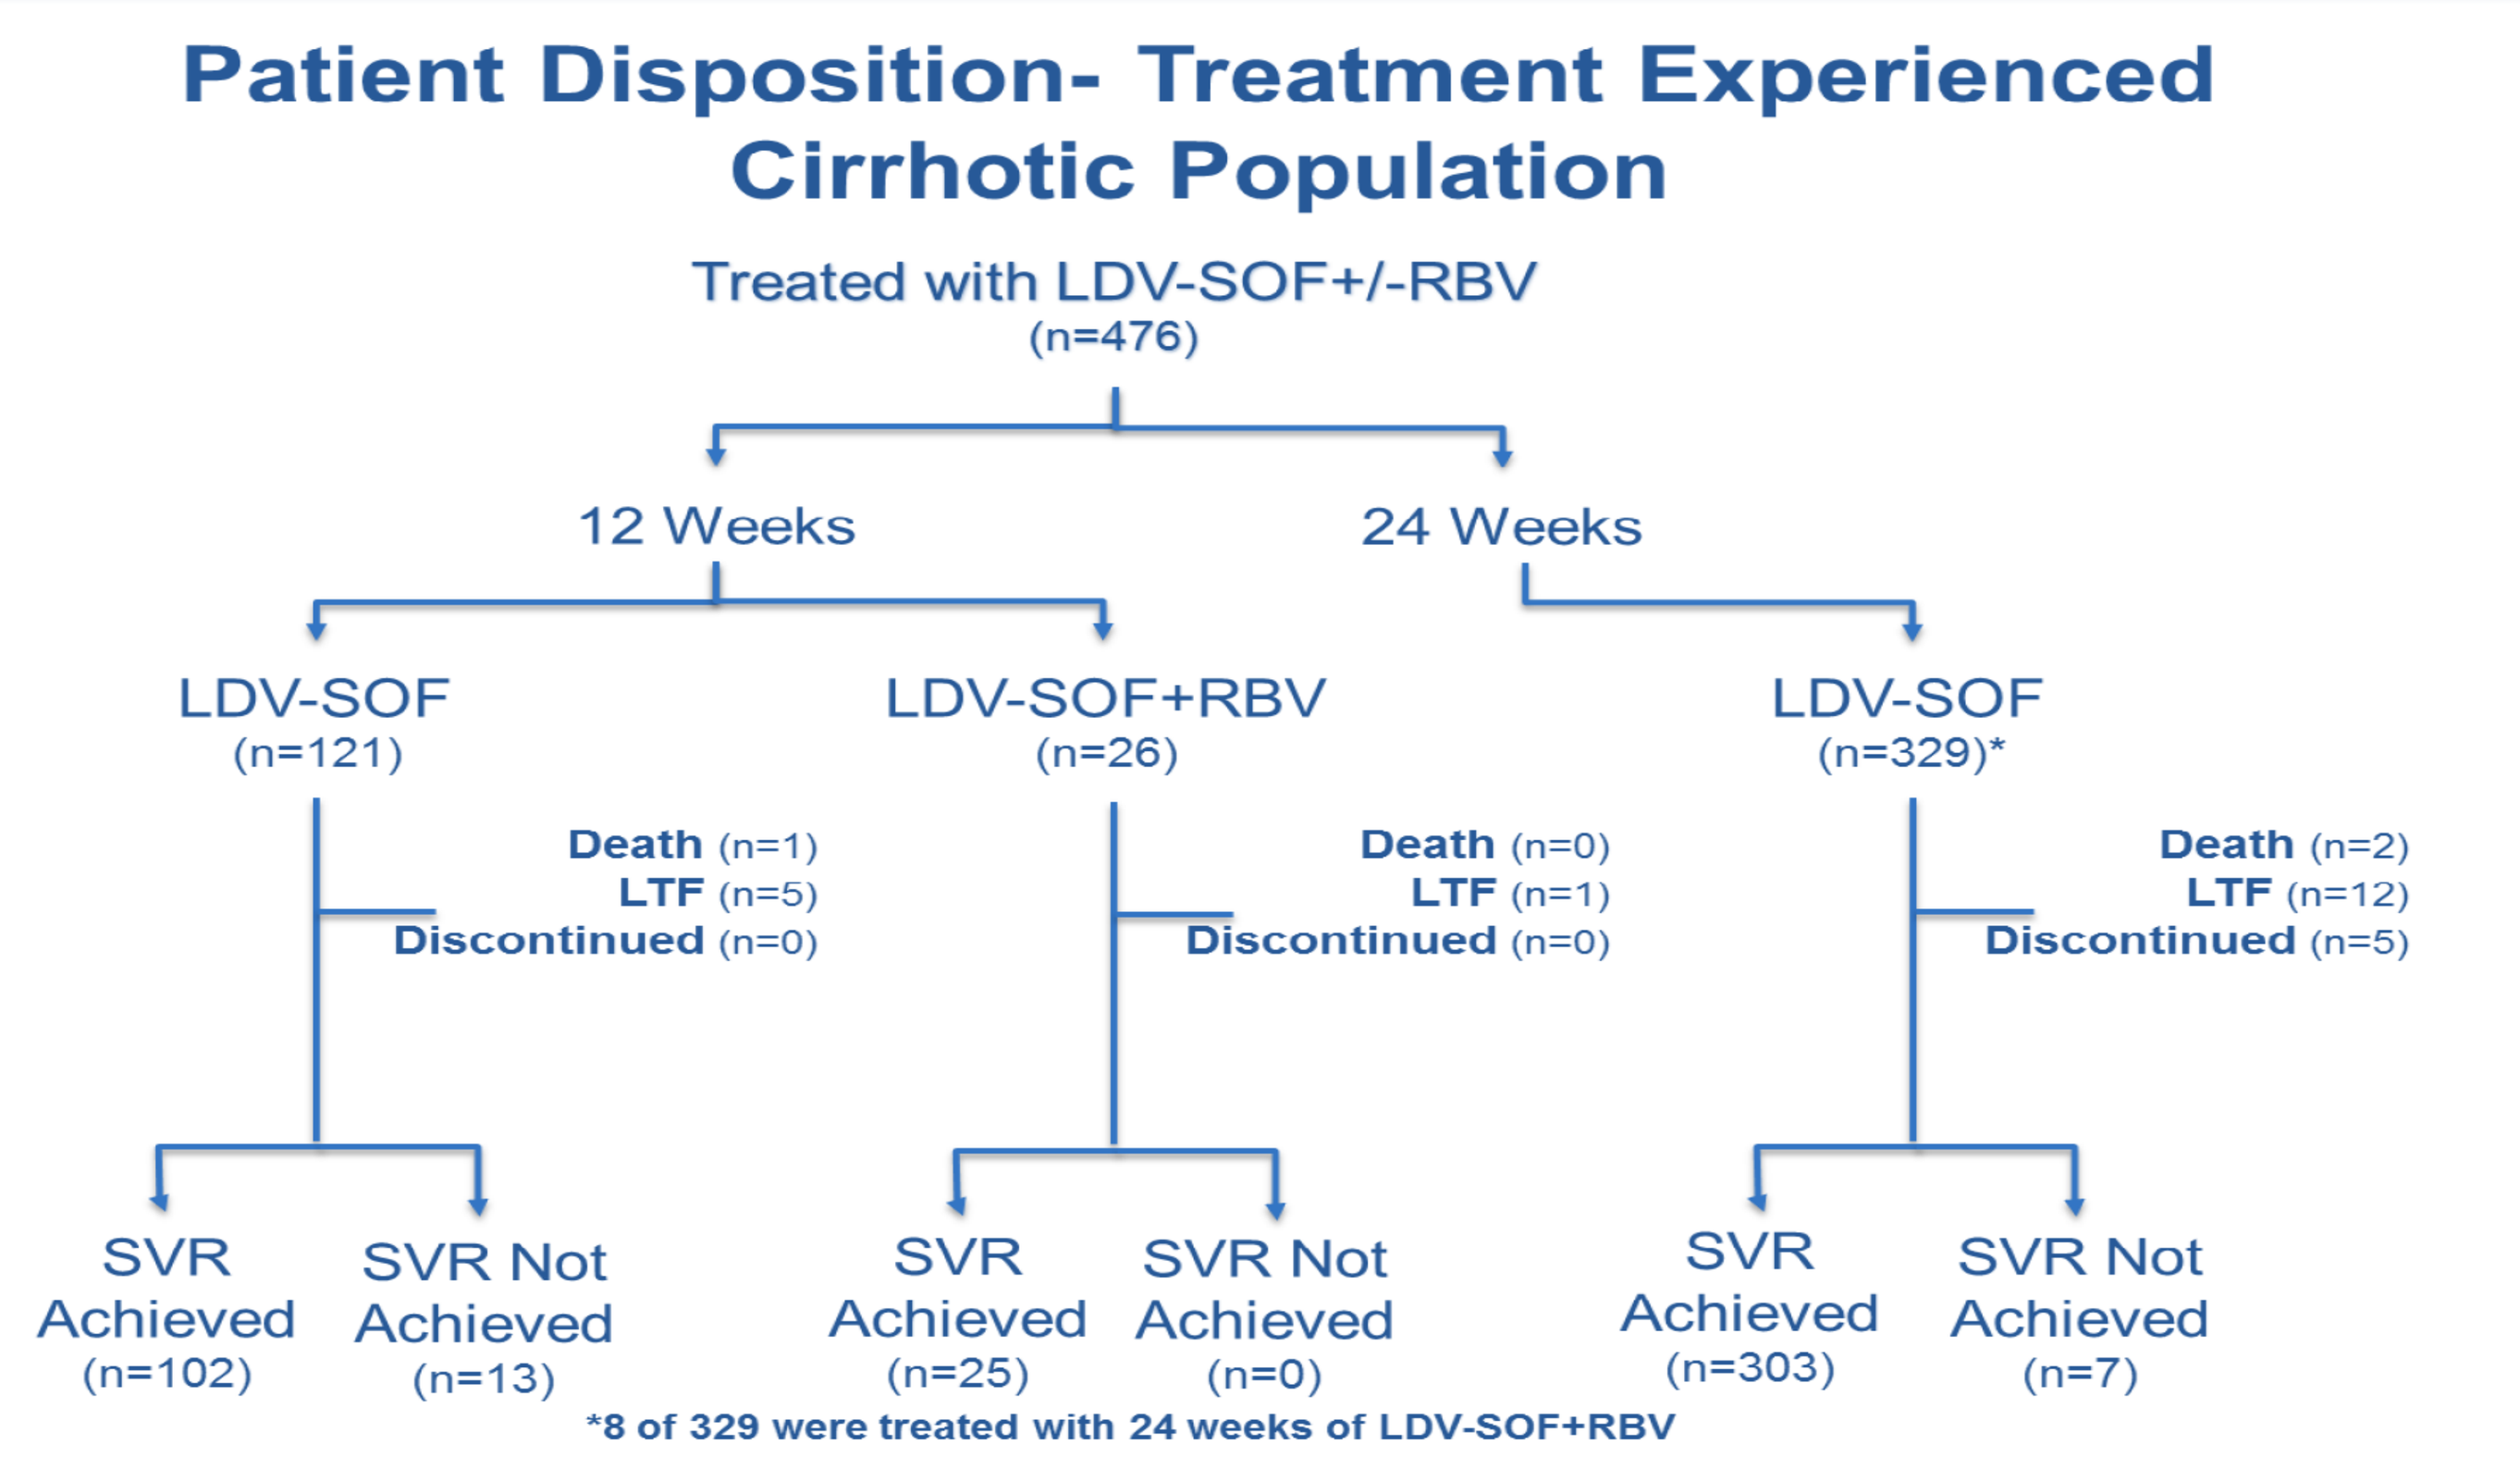
Patient Disposition- Treatment Experienced Cirrhotic Population
Treated with LDV-SOF+/-RBV
(n=476)
12 Weeks
24 Weeks
LDV-SOF
(n=121)
LDV-SOF+RBV
(n=26)
LDV-SOF
(n=329)*
Death (n=1)
LTF (n=5)
Discontinued (n=0)
Death (n=0)
LTF (n=1)
Discontinued (n=0)
Death (n=2)
LTF (n=12)
Discontinued (n=5)
SVR Achieved (n=102)
SVR Not Achieved (n=13)
SVR Achieved (n=25)
SVR Not Achieved (n=0)
SVR Achieved (n=303)
SVR Not Achieved (n=7)
*8 of 329 were treated with 24 weeks of LDV-SOF+RBV

5. BASELINE CHARACTERISTICS

Patients on a 12 or 24 week regimen, n = 476

Patient Demographics and Clinical Characteristics at Baseline

Group - no. (%)	LDV-SOF 12 Week n= 121	LDV-SOF + RBV 12 Week n= 26	*LDV-SOF 24 Week n= 329
Academic Practice – no. (%)	50 (41%)	21 (81%)	189 (57%)
Age - mean (range)	61 (29-88)	58 (40-71)	61 (26-84)
Male - no. (%)	73 (60%)	16 (62%)	230 (70%)
Patient Ethnicity - no. (%)			
Asian	12 (10%)	1 (4%)	4 (1%)
Black	17 (14%)	1 (4%)	28 (8%)
Hispanic/Latin	13 (11%)	1 (4%)	26 (8%)
Native Hawaiian or Other Pacific Islander	5 (4%)	0 (0%)	0 (0%)
White	44 (36%)	19 (73%)	209 (64%)
Other	1 (1%)	0 (0%)	3 (1%)
Unknown	29 (24%)	4 (15%)	59 (18%)
Baseline Labs			
ALT - mean (s.d.)	69 (61), n=111	92 (59), n=25	86 (59), n=314
AST - mean (s.d.)	72 (52), n=110	92 (52), n=25	86 (53), n=314
Hb - mean (s.d.)	13 (2), n=106	15 (1), n=24	14 (3), n=321
Platelets <100 K/ml - no. (%)	26 (25%)	5 (21%)	129 (41%)
Platelets >=100 K/ml - no. (%)	76 (75%)	19 (79%)	183 (59%)
Genotype - no. (%)			
1a	75 (62%)	18 (69%)	226 (69%)
1b	41 (34%)	7 (27%)	82 (25%)
1Mixed	1 (1%)	0 (0%)	0 (0%)
1Unknown	4 (3%)	1 (4%)	21 (6%)
Initial Viral Load - no. (%)			
<800K IU/ml	37 (31%)	13 (50%)	107 (33%)
800K<2MM IU/ml	35 (29%)	2 (8%)	90 (27%)
2MM<6MM IU/ml	27 (22%)	6 (23%)	86 (26%)
6MM+ IU/ml	21 (17%)	5 (19%)	41 (12%)
Unknown	1 (1%)	0 (0%)	5 (2%)
Prior Treatment - no. (%)			
PEG + RBV	41 (53%)	9 (36%)	181 (55%)
BOC + PEG + RBV	7 (9%)	0 (0%)	24 (7%)
TVR + PEG + RBV	8 (10%)	9 (36%)	56 (17%)
SMV + SOF + RBV	0 (0%)	0 (0%)	3 (1%)
SMV + SOF	2 (3%)	3 (12%)	27 (8%)
SOF + PEG + RBV	2 (3%)	0 (0%)	8 (3%)
SOF + RBV	5 (6%)	3 (12%)	6 (2%)
LDV-SOF	1 (1%)	0 (0%)	0 (0%)
Prior Regimen Unknown	12 (15%)	1 (4%)	24 (7%)
Prior Response - no. (%)			
Discontinued	7 (6%)	0 (0%)	20 (6%)
Null Responder	36 (30%)	11 (42%)	128 (39%)
Responder Relapse	22 (18%)	12 (46%)	126 (38%)
Unknown	56 (46%)	3 (12%)	56 (17%)

This study was funded by Gilead Sciences, Inc.

One or more of the authors have served as a consultant or scientific advisor for AbbVie, Achillion Pharmaceuticals, BioCryst, Biotica, Bristol-Myers Squibb, Enanta, Evidera, Gilead Sciences, Inc, Idenix Pharmaceuticals, Inc, Janssen Therapeutics, Merck & Co, Inc, Novartis, Regulus, Santaris Pharmaceuticals, Tekmira Pharmaceuticals, Theravance, Trio Health, Vertex Pharmaceuticals; received grant or research support from AbbVie, Beckman, Boehringer Ingelheim, Bristol-Myers Squibb, Conatus Pharmaceuticals, Inc, Gilead Sciences, Inc, Idenix Pharmaceuticals, Inc, Ikaria, Intercept, Janssen Therapeutics, MassBiologics, Merck & Co, Inc, Novartis, Presidio, Roche, Santaris Pharmaceuticals, Theravance, Vertex Pharmaceuticals; and/or received speaker honoraria from AbbVie, Bayer, Bristol-Myers Squibb, Gilead Sciences, Inc, Janssen Therapeutics, Kadmon, Merck & Co, Inc, Roche, Salix, Vertex Pharmaceuticals.

6. SVR12 RATES

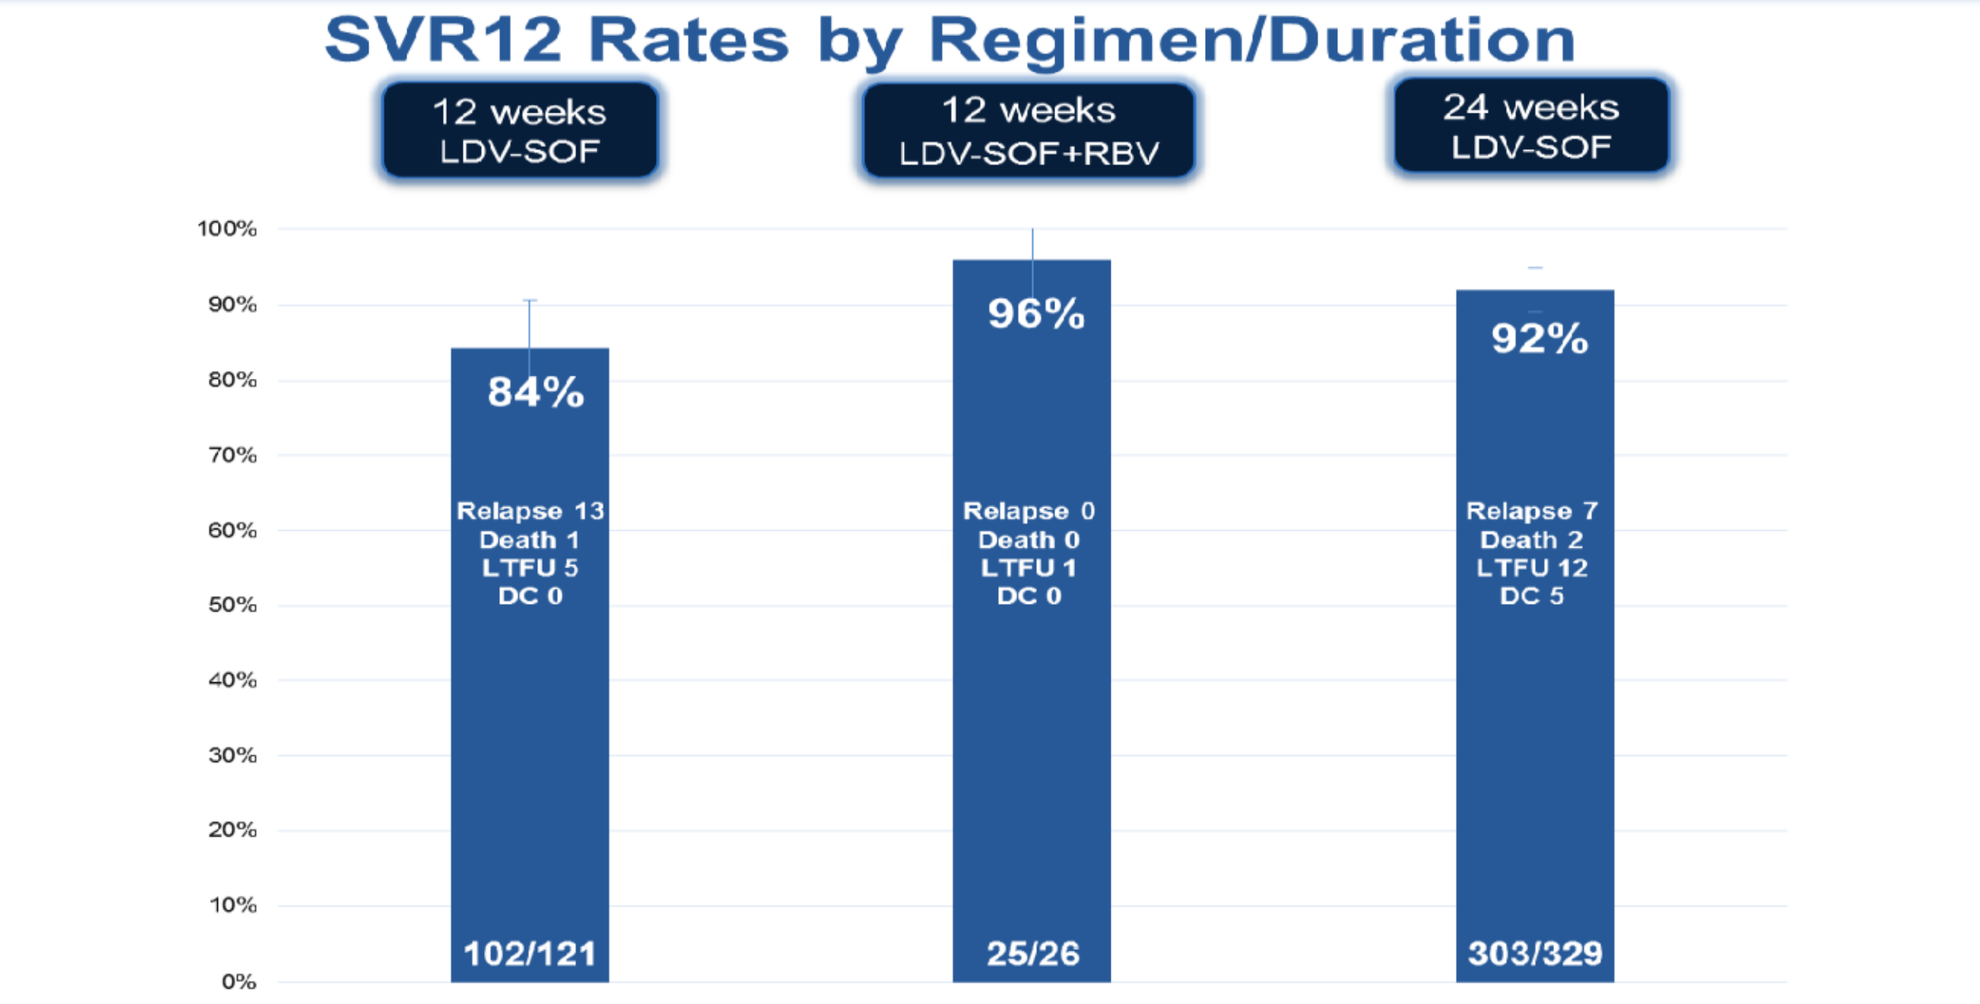
SVR12 Rates by Regimen/Duration
12 weeks LDV-SOF
12 weeks LDV-SOF+RBV
24 weeks LDV-SOF
84%
96%
92%
Relapse 13 Death 1 LTFU 5 DC 0
Relapse 0 Death 0 LTFU 1 DC 0
Relapse 7 Death 2 LTFU 12 DC 5
102/121
25/26
303/329

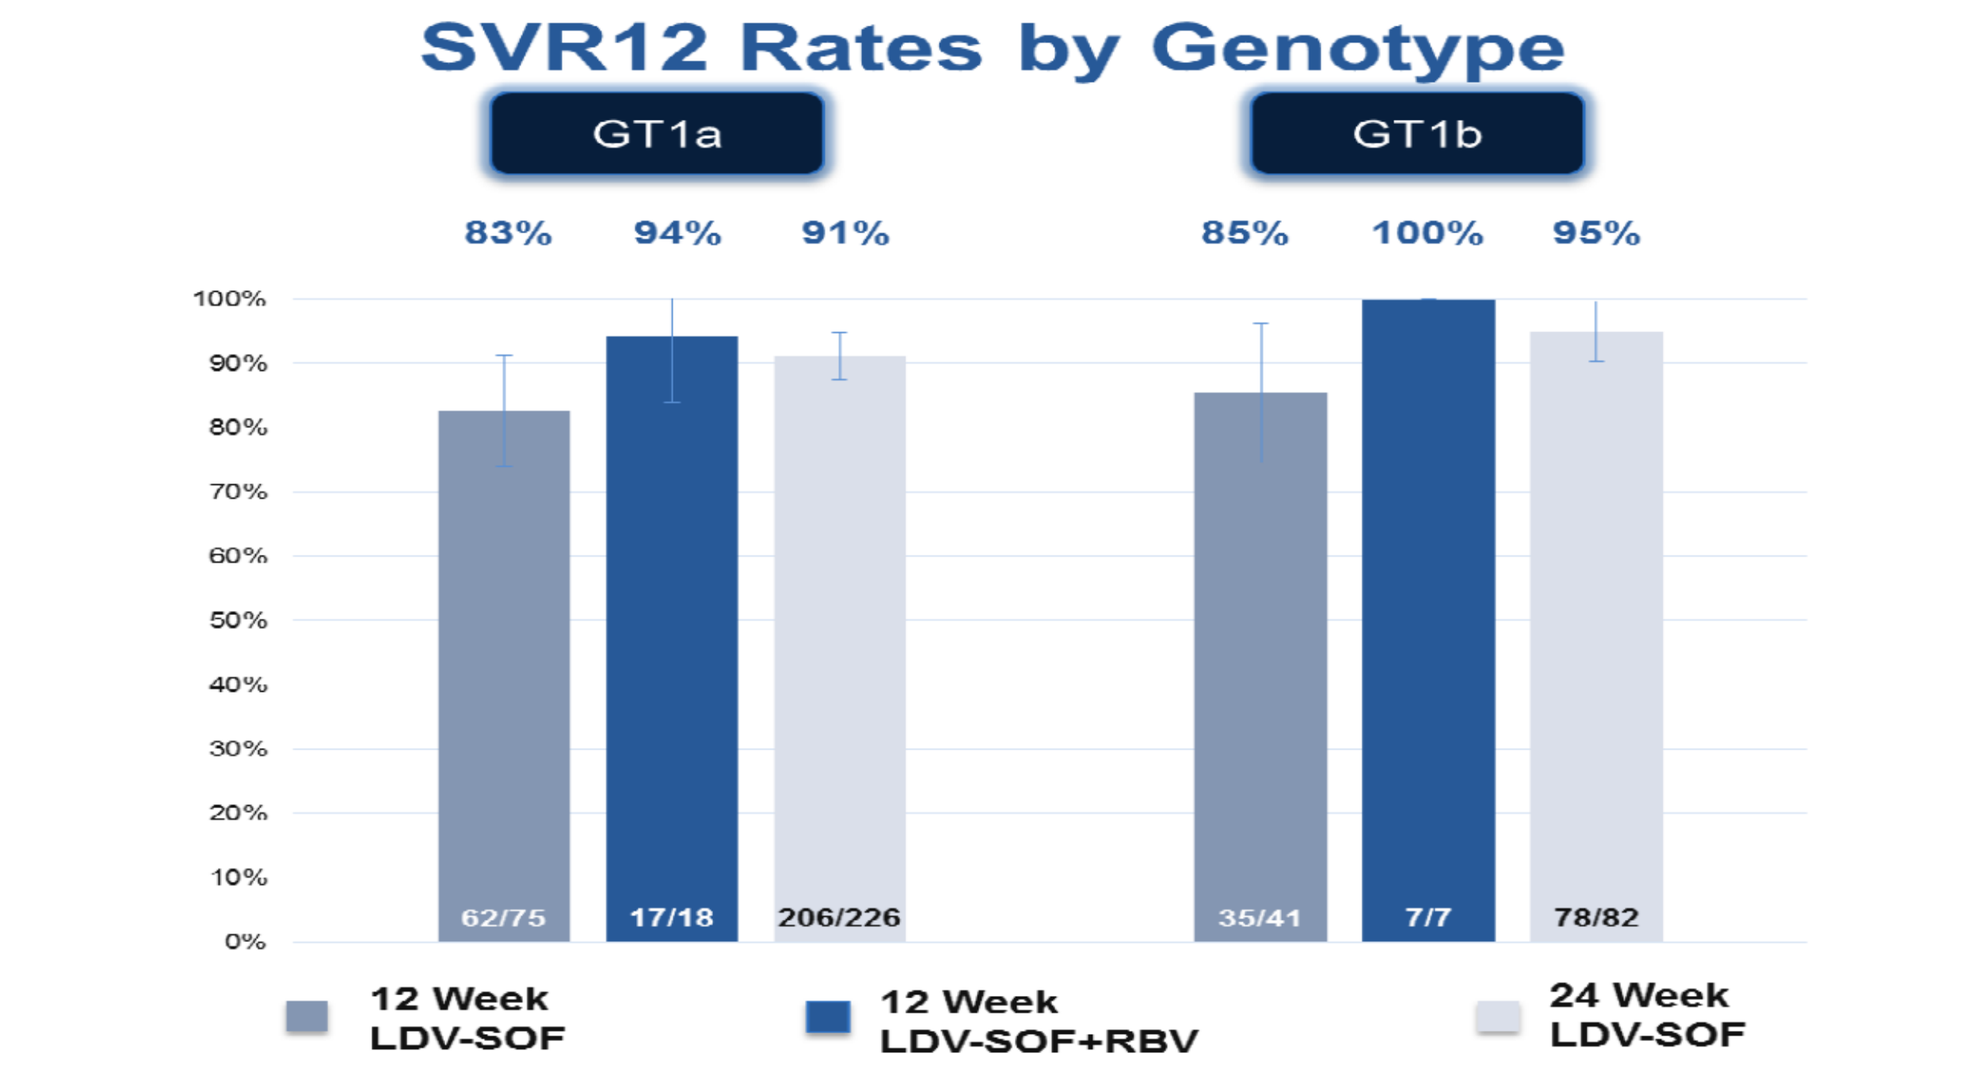
SVR12 Rates by Genotype
GT1a
GT1b
83% 94% 91%
85% 100% 95%
62/75
17/18
206/226
35/41
7/7
78/82
12 Week LDV-SOF
12 Week LDV-SOF+RBV
24 Week LDV-SOF

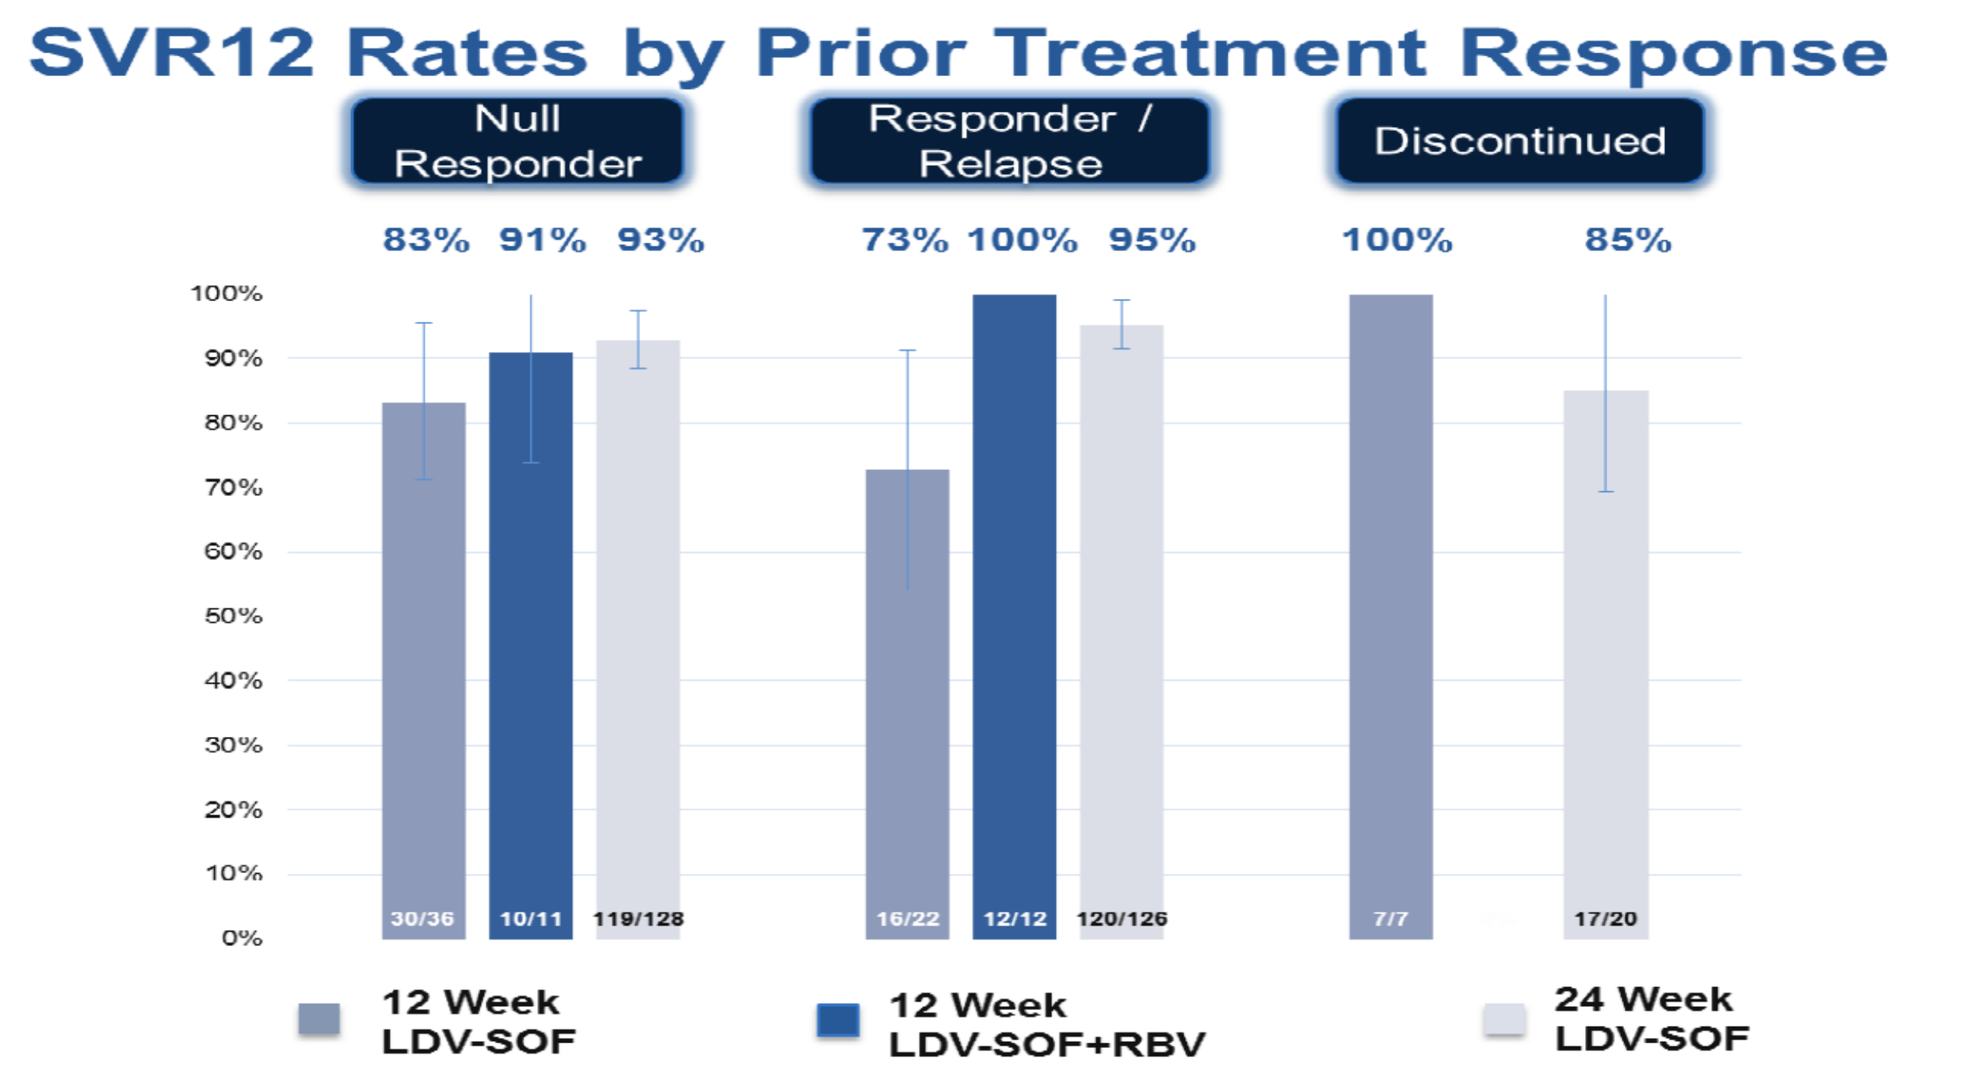
SVR12 Rates by Prior Treatment Response
Null Responder
Responder / Relapse
Discontinued
83% 91% 93%
73% 100% 95%
100% 85%
30/36
10/11
119/128
16/22
12/12
120/126
7/7
17/20
12 Week LDV-SOF
12 Week LDV-SOF+RBV
24 Week LDV-SOF

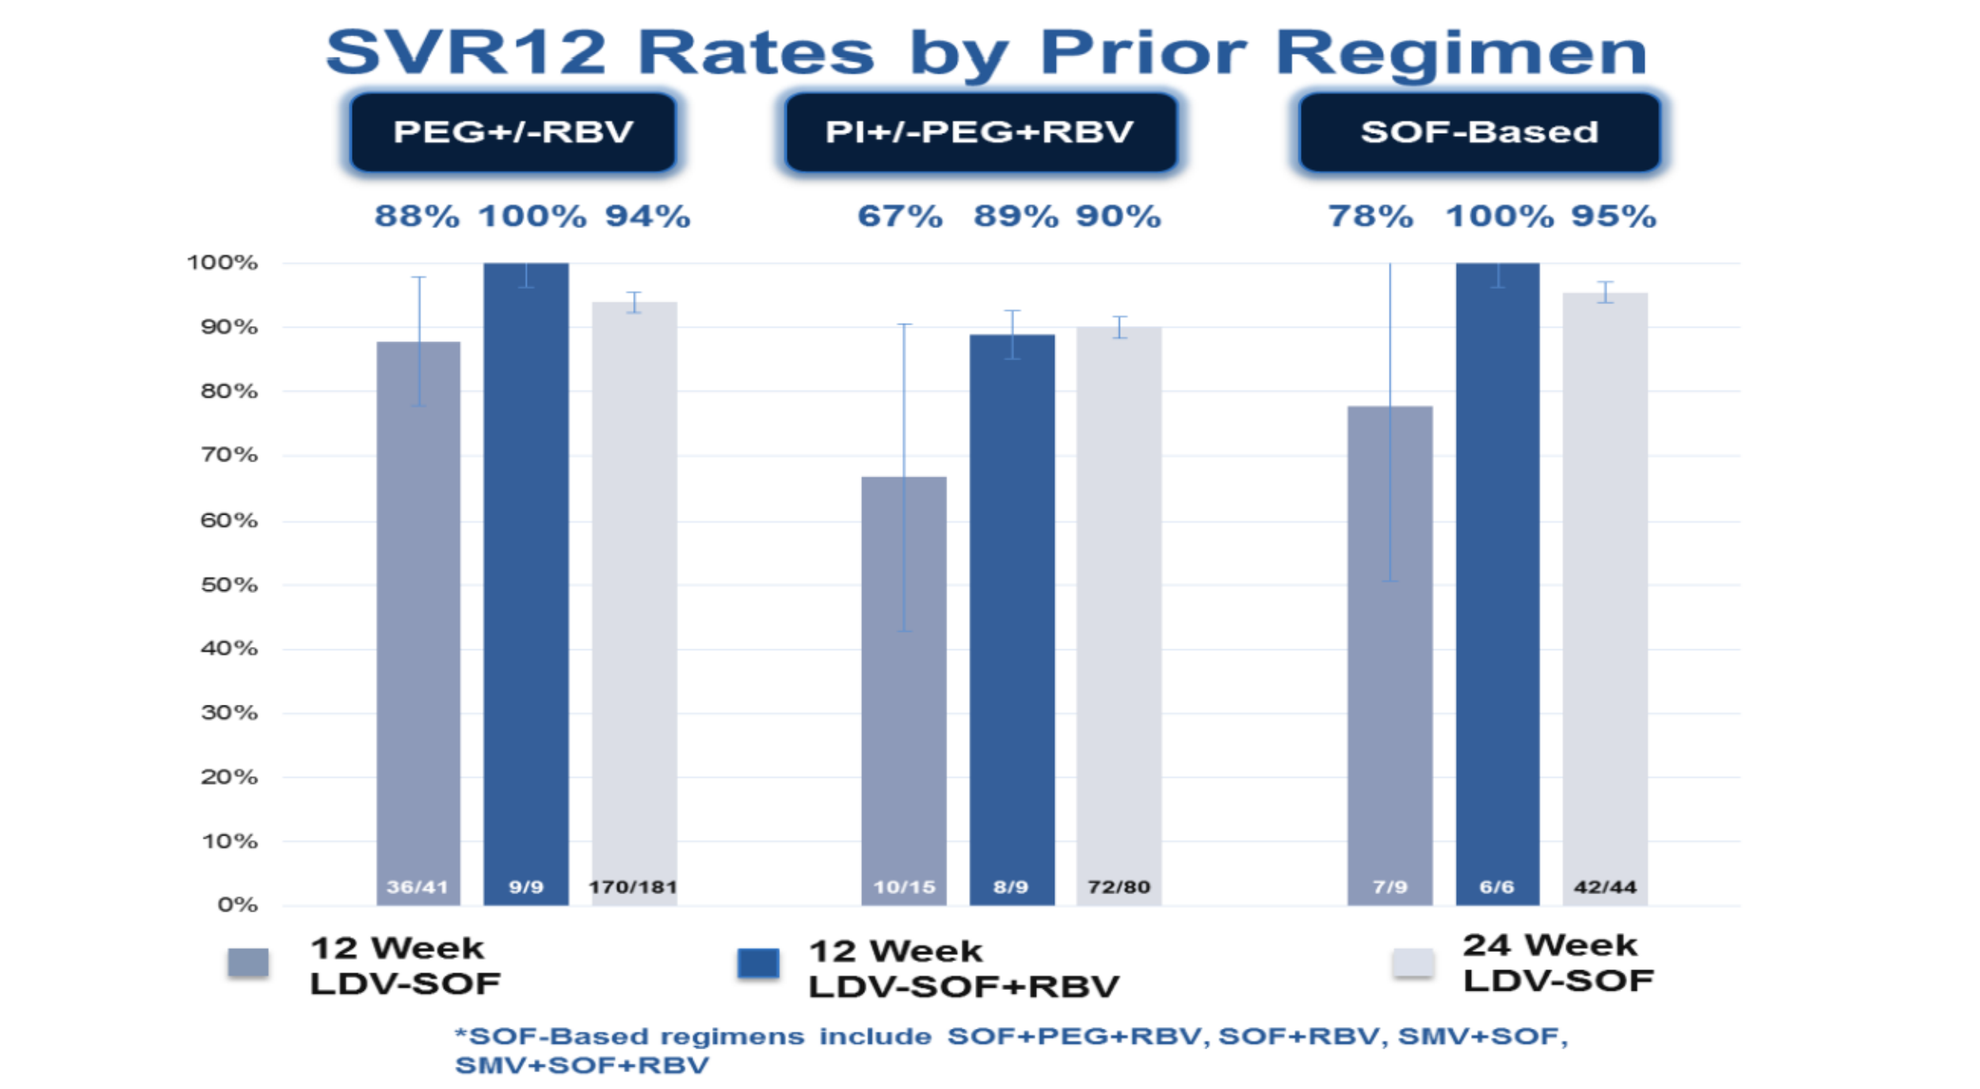
SVR12 Rates by Prior Regimen
PEG+/-RBV
PI+/-PEG+RBV
SOF-Based
88% 100% 94%
67% 89% 90%
78% 100% 95%
36/41
9/9
170/181
10/15
8/9
72/80
7/9
6/6
42/44
12 Week LDV-SOF
12 Week LDV-SOF+RBV
24 Week LDV-SOF
*SOF-Based regimens include SOF+PEG+RBV, SOF+RBV, SMV+SOF, SMV+SOF+RBV

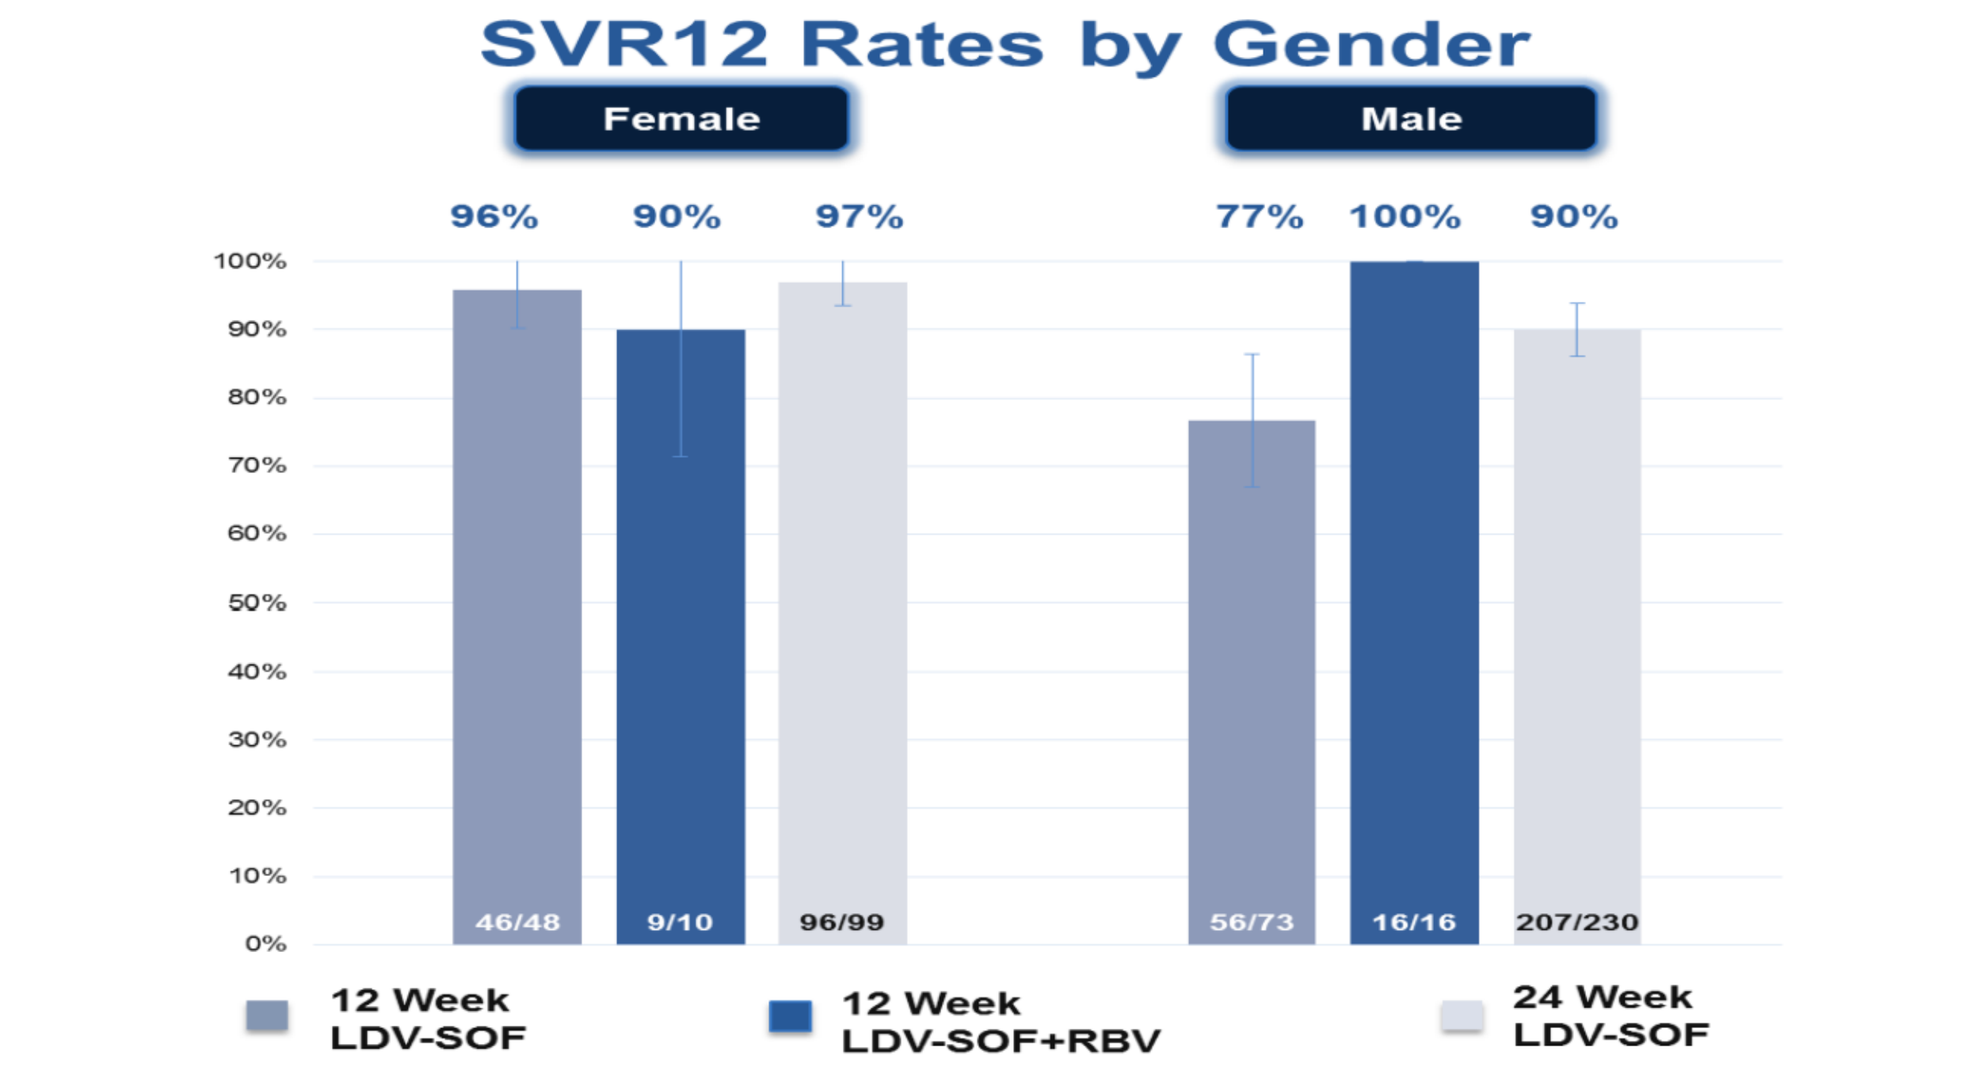
SVR12 Rates by Gender
Female
Male
96% 90% 97%
77% 100% 90%
46/48
9/10
96/99
56/73
16/16
207/230
12 Week LDV-SOF
12 Week LDV-SOF+RBV
24 Week LDV-SOF

Numerator = SVR Achieved; Denominator = Intent to Treat (ITT) Whiskers = 95% confidence intervals.

7. PREDICTORS OF RESPONSE

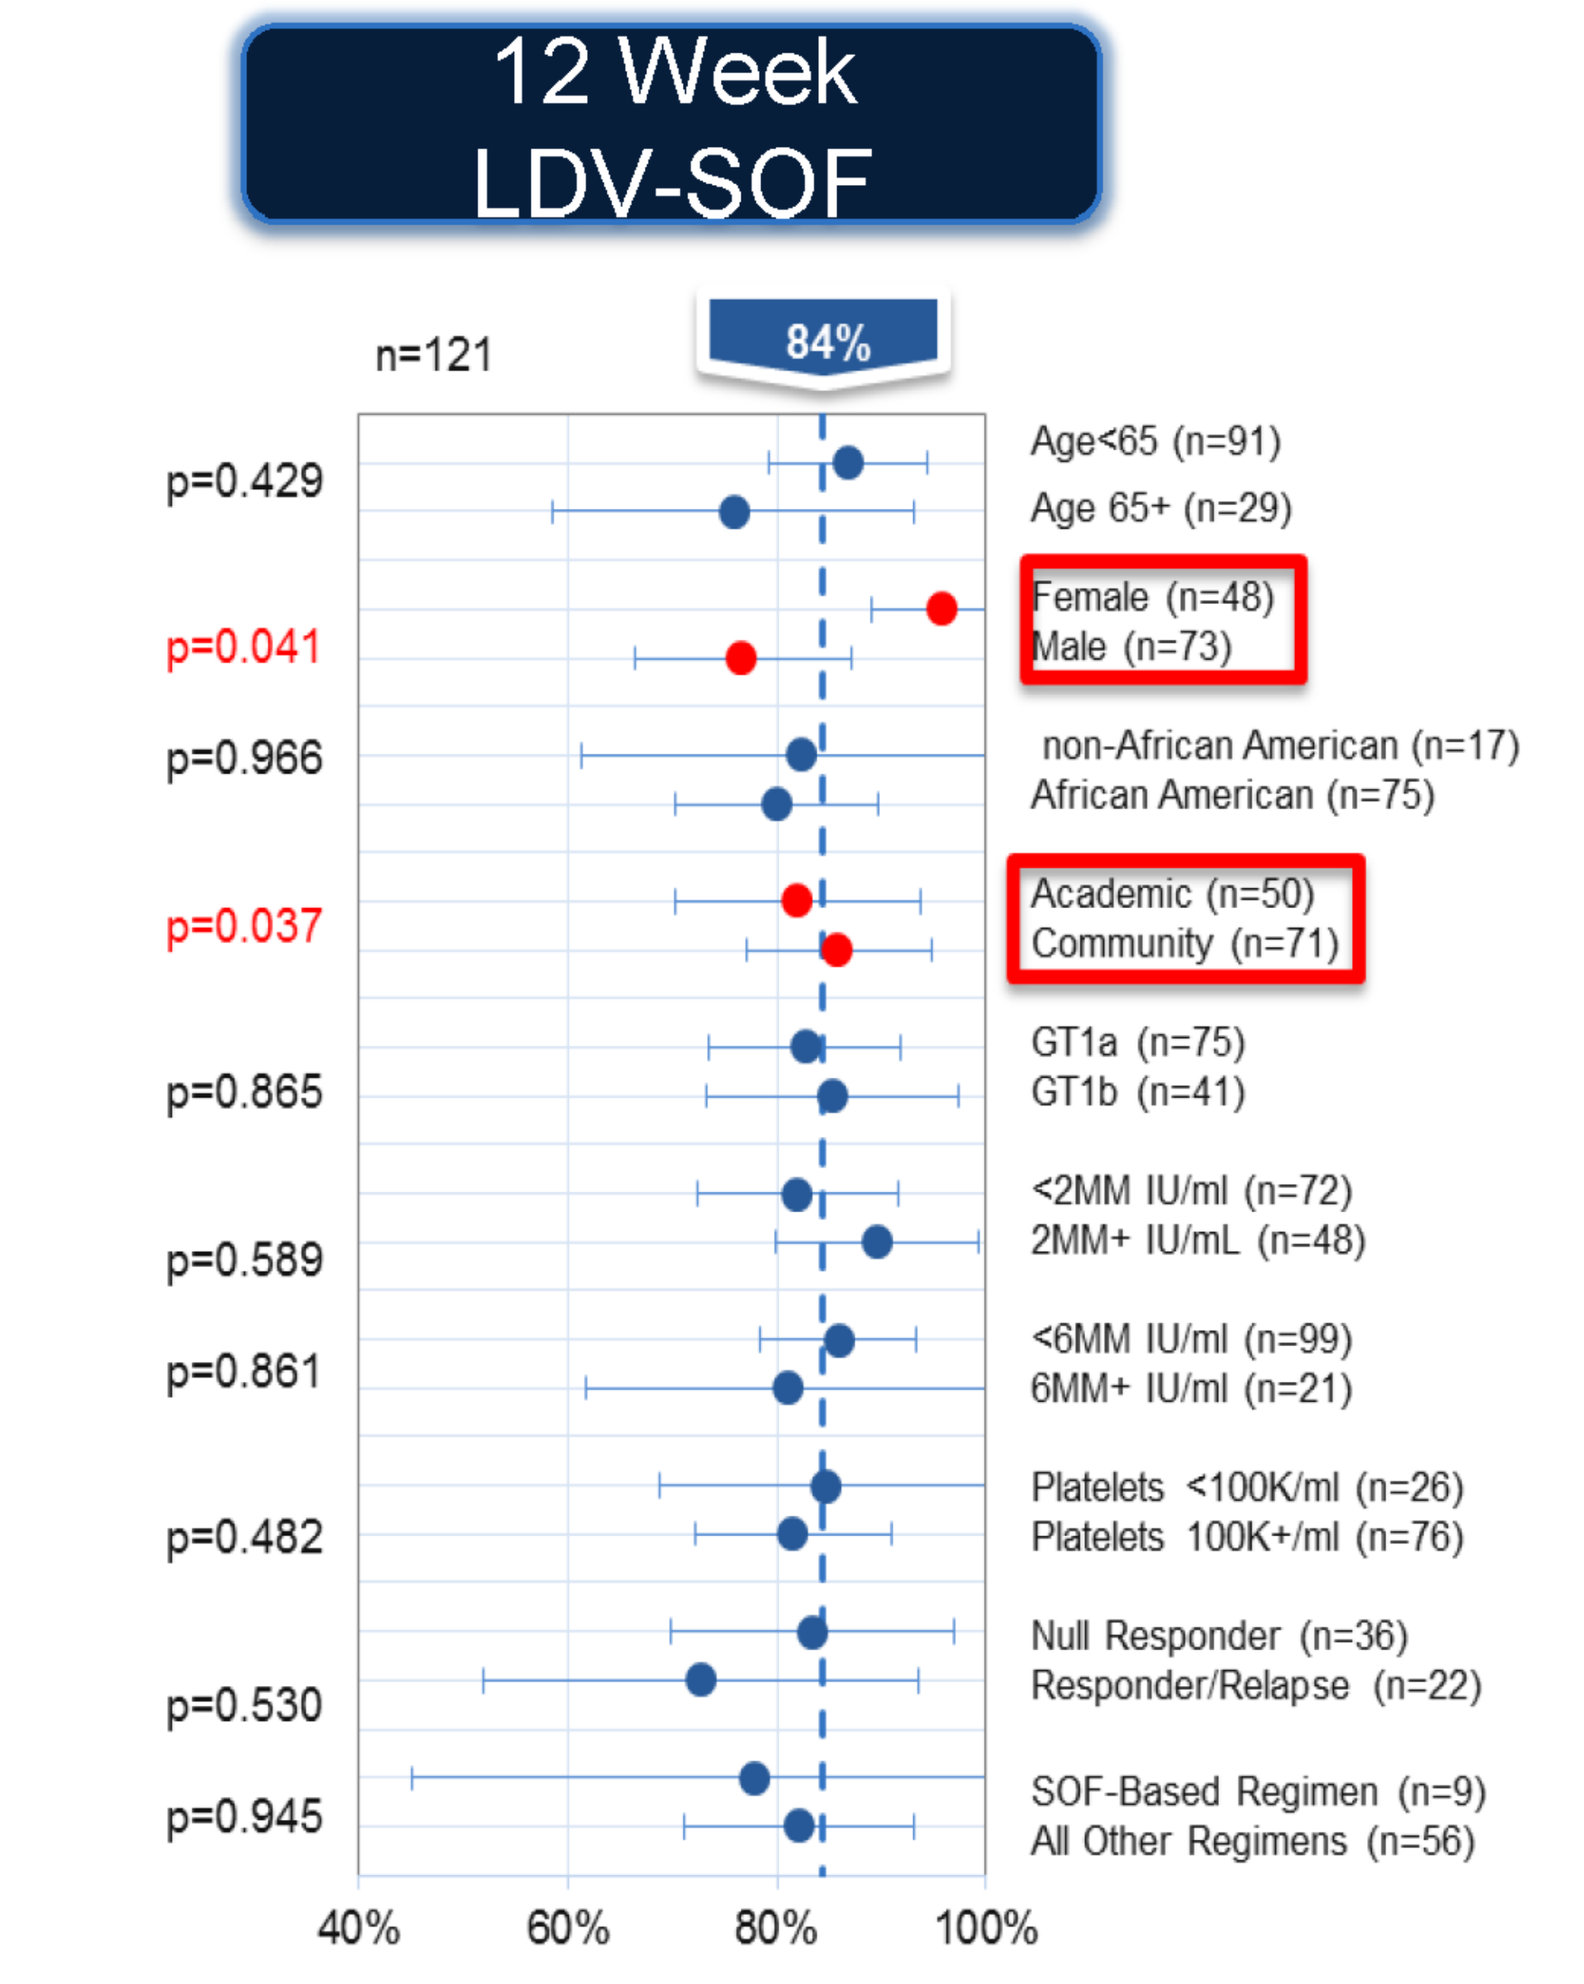
12 Week LDV-SOF
n=121
84%
p=0.429 Age<65 (n=91) Age 65+ (n=29)
p=0.041 Female (n=48) Male (n=73)
p=0.966 non-African American (n=17) African American (n=75)
p=0.037 Academic (n=50) Community (n=71)
p=0.865 GT1a (n=75) GT1b (n=41)
p=0.589 <2MM IU/ml (n=72) 2MM+ IU/mL (n=48)
p=0.861 <6MM IU/ml (n=99) 6MM+ IU/ml (n=21)
p=0.482 Platelets <100K/ml (n=26) Platelets 100K+/ml (n=76)
p=0.530 Null Responder (n=36) Responder/Relapse (n=22)
p=0.945 SOF-Based Regimen (n=9) All Other Regimens (n=56)
40% 60% 80% 100%

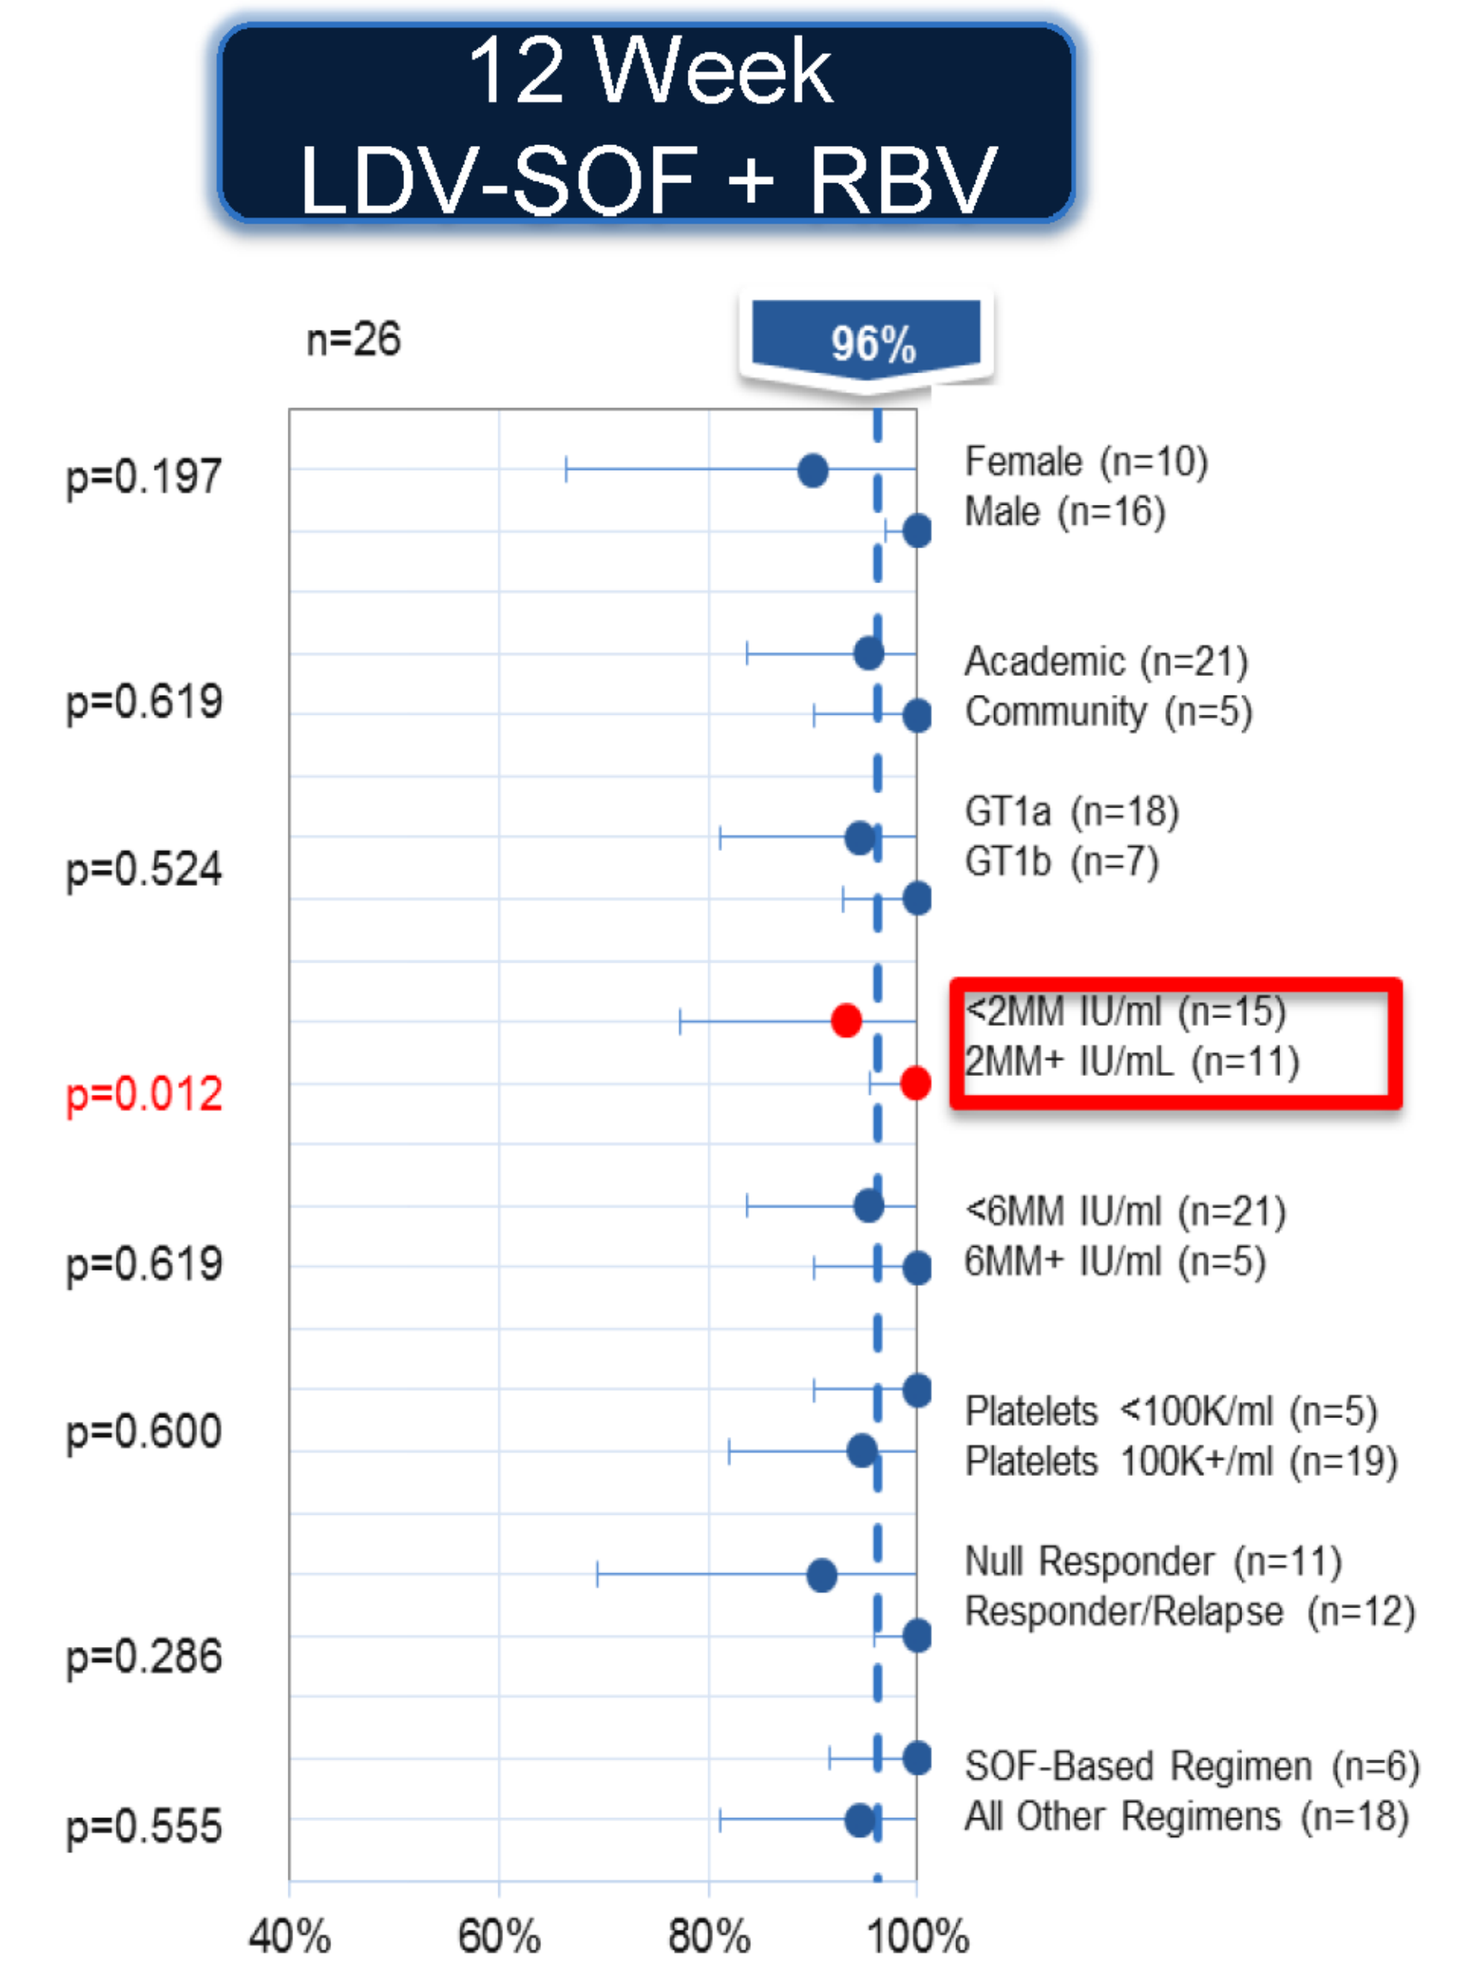
12 Week LDV-SOF + RBV
n=26
96%
p=0.197 Female (n=10) Male (n=16)
p=0.619 Academic (n=21) Community (n=5)
p=0.524 GT1a (n=18) GT1b (n=7)
p=0.012 <2MM IU/ml (n=15) 2MM+ IU/mL (n=11)
p=0.619 <6MM IU/ml (n=21) 6MM+ IU/ml (n=5)
p=0.600 Platelets <100K/ml (n=5) Platelets 100K+/ml (n=19)
p=0.286 Null Responder (n=11) Responder/Relapse (n=12)
p=0.555 SOF-Based Regimen (n=6) All Other Regimens (n=18)
40% 60% 80% 100%

*Age excluded from LDV-SOF+RBV plot due to n<5 in 65+ group. Ethnicity excluded from LDV-SOF+RBV plot due to n<5 in African American group

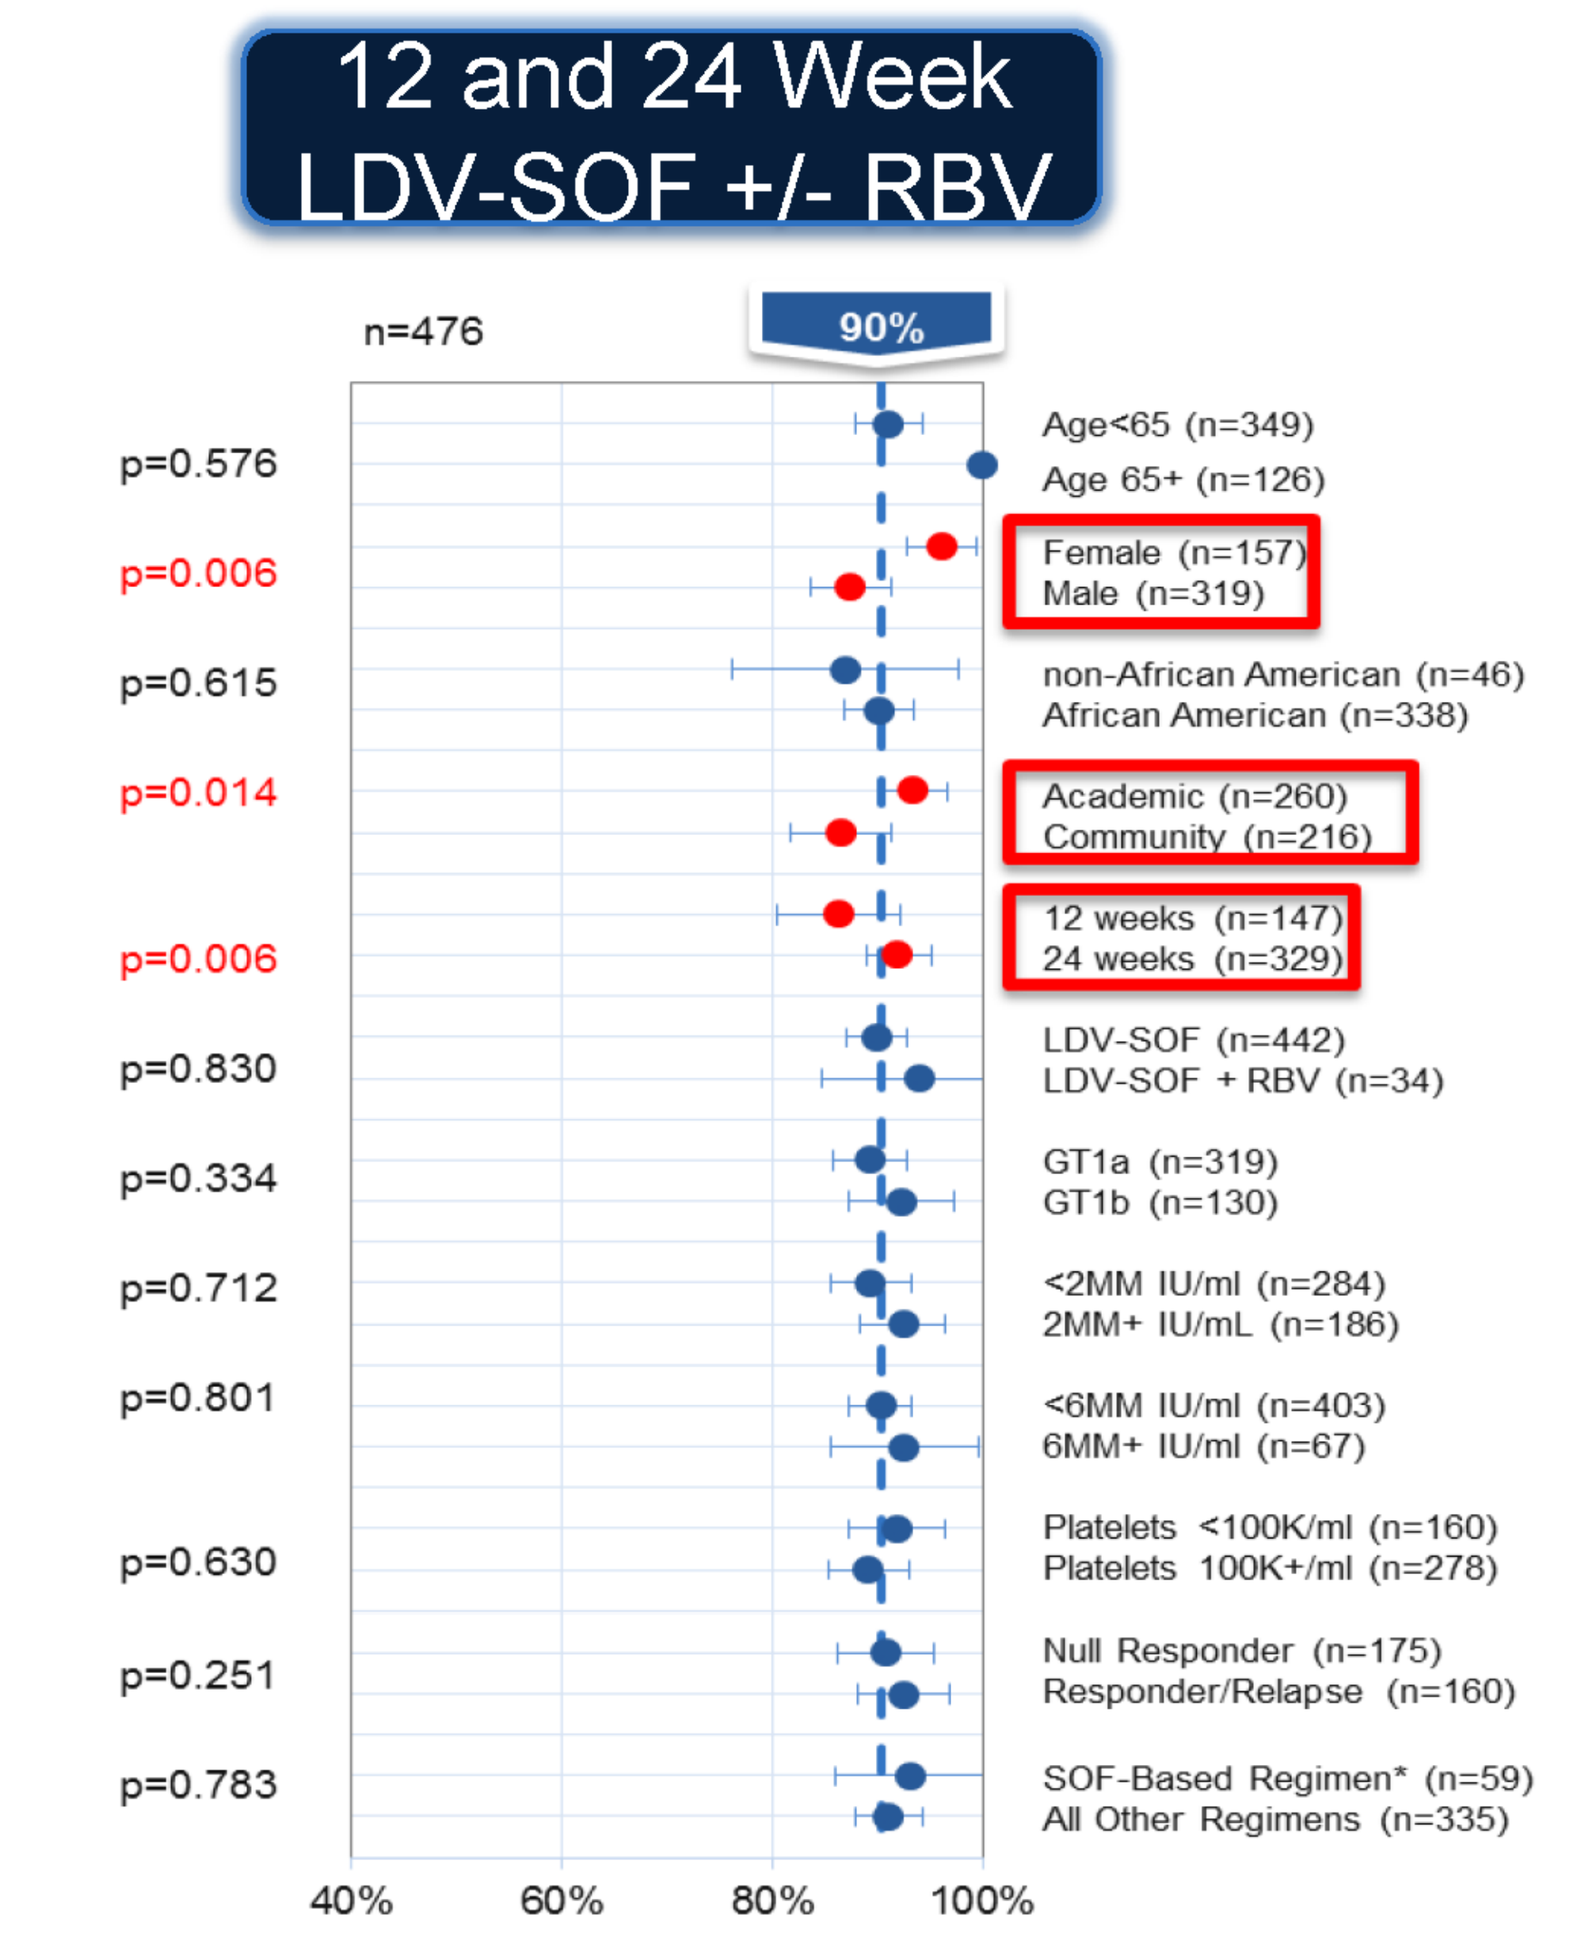
12 and 24 Week LDV-SOF +/- RBV
n=476
90%
p=0.576 Age<65 (n=349) Age 65+ (n=126)
p=0.006 Female (n=157) Male (n=319)
p=0.615 non-African American (n=46) African American (n=338)
p=0.014 Academic (n=260) Community (n=216)
p=0.006 12 weeks (n=147) 24 weeks (n=329)
p=0.830 LDV-SOF (n=442) LDV-SOF + RBV (n=34)
p=0.334 GT1a (n=319) GT1b (n=130)
p=0.712 <2MM IU/ml (n=284) 2MM+ IU/mL (n=186)
p=0.801 <6MM IU/ml (n=403) 6MM+ IU/ml (n=67)
p=0.630 Platelets <100K/ml (n=160) Platelets 100K+/ml (n=278)
p=0.251 Null Responder (n=175) Responder/Relapse (n=160)
p=0.783 SOF-Based Regimen* (n=59) All Other Regimens (n=335)
40% 60% 80% 100%

*SOF-Based regimens include SOF+PEG+RBV, SOF+RBV, SMV+SOF, SMV+SOF+RBV

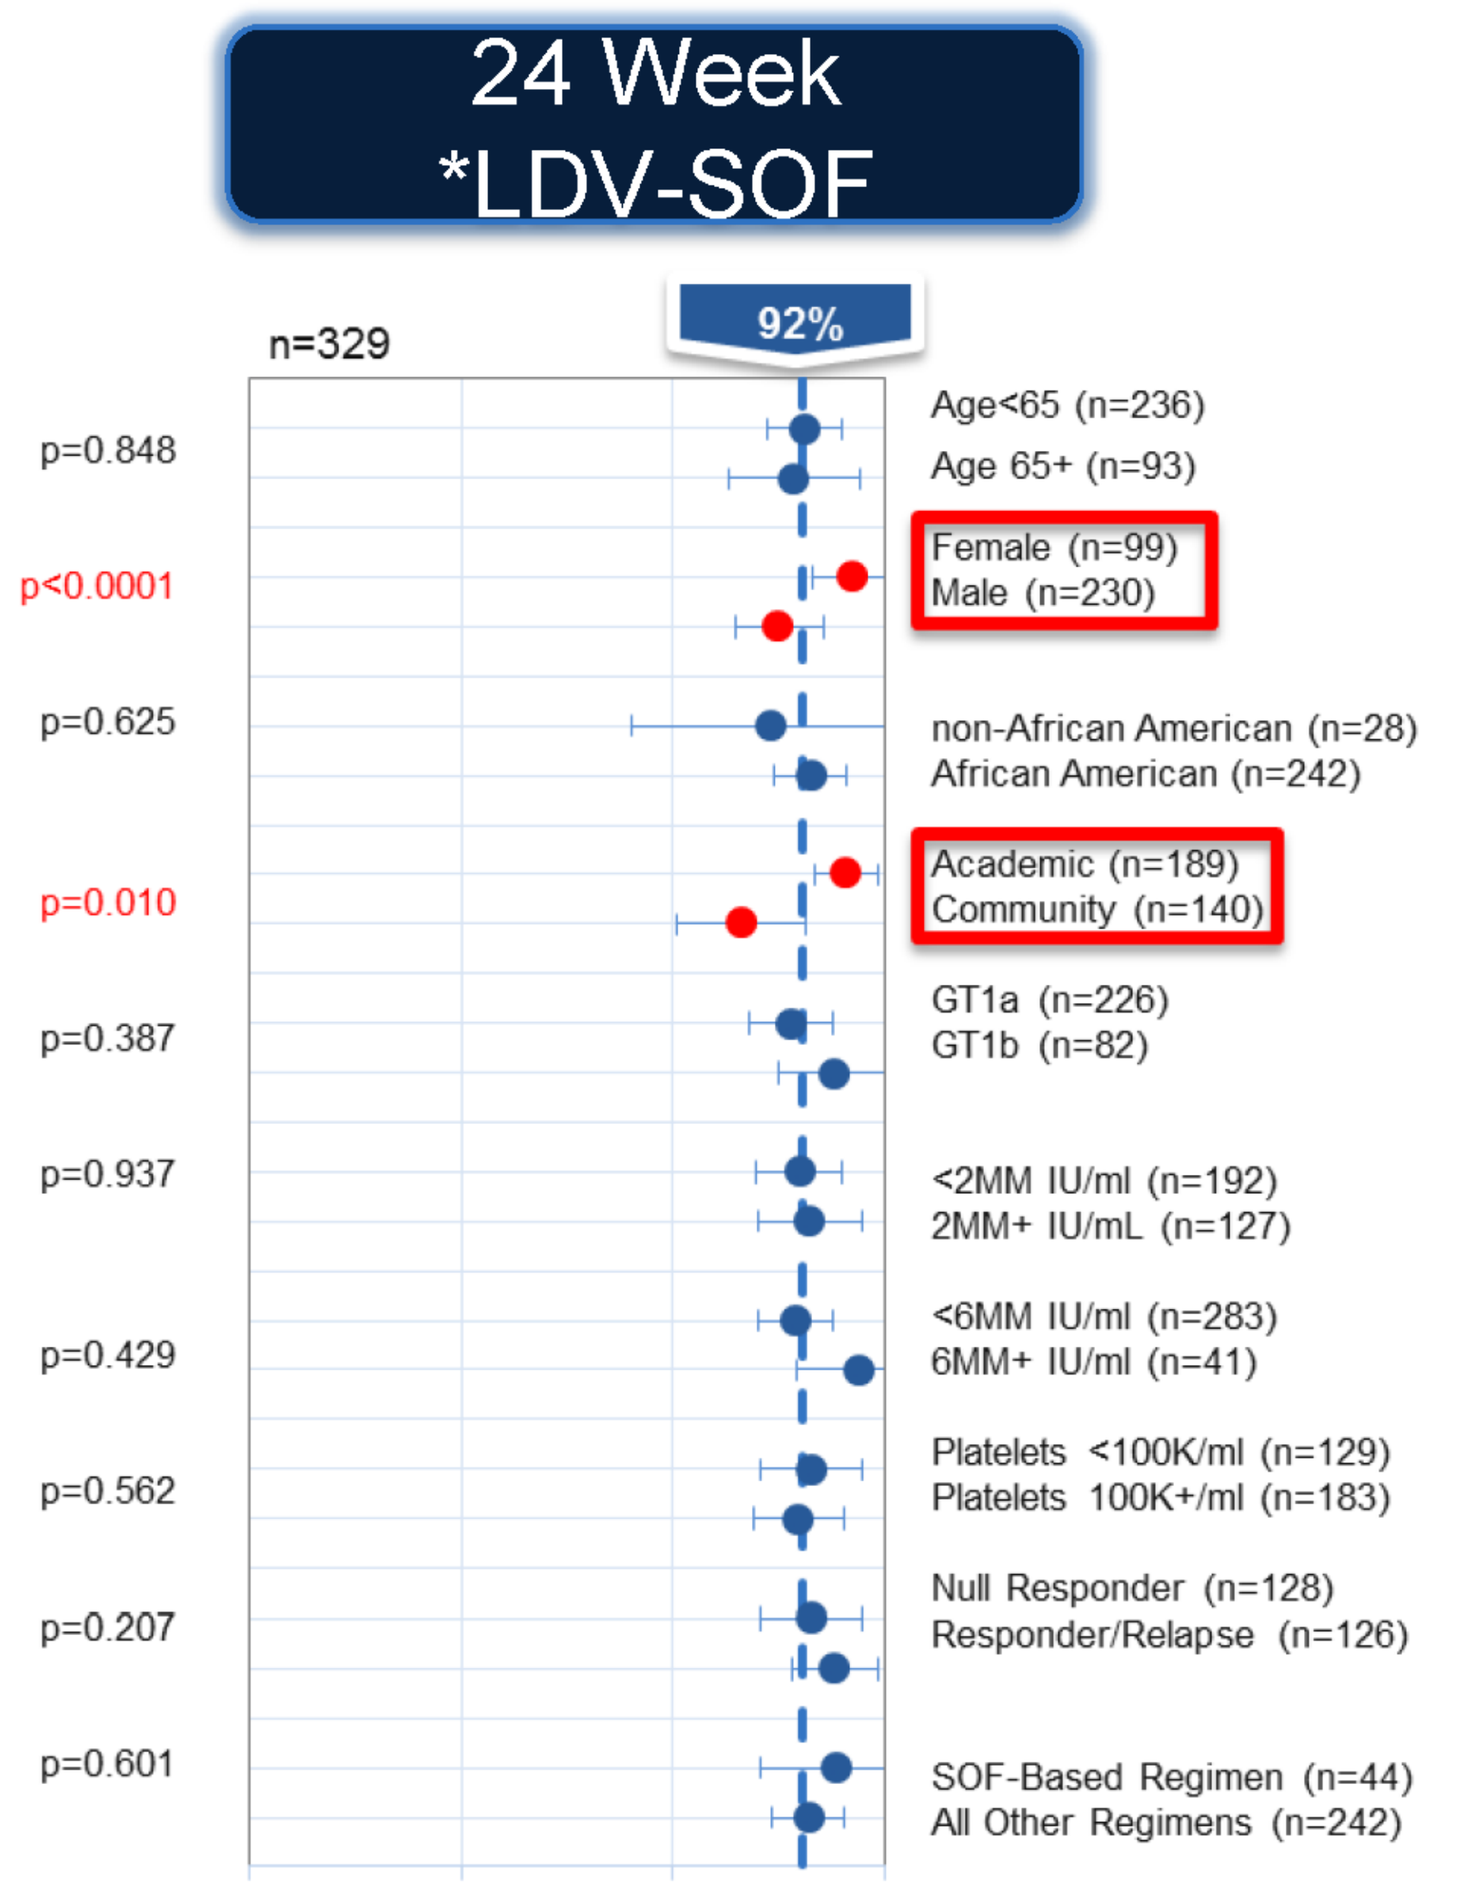
24 Week *LDV-SOF
n=329
92%
p=0.848 Age<65 (n=236) Age 65+ (n=93)
p<0.0001 Female (n=99) Male (n=230)
p=0.625 non-African American (n=28) African American (n=242)
p=0.010 Academic (n=189) Community (n=140)
p=0.387 GT1a (n=226) GT1b (n=82)
p=0.937 <2MM IU/ml (n=192) 2MM+ IU/mL (n=127)
p=0.429 <6MM IU/ml (n=283) 6MM+ IU/ml (n=41)
p=0.562 Platelets <100K/ml (n=129) Platelets 100K+/ml (n=183)
p=0.207 Null Responder (n=128) Responder/Relapse (n=126)
p=0.601 SOF-Based Regimen (n=44) All Other Regimens (n=242)
40% 60% 80% 100%

Circles indicate SVR12, whiskers = 95% confidence intervals. Rates that are significantly different are indicated by red circles.

8. SUMMARY

From a data collection of 476 treatment-experienced patients with cirrhosis, genotype 1 Hepatitis C, SVR 12 for 12 week LDV-SOF, 12 week LDV-SOF+RBV, and 24 week LDV-SOF regimens were 84%, 96%, and 92% respectively. In real world, 12 week LDV-SOF+RBV (96%) was similar to 24 week LDV-SOF (92%).

Overall SVR rate was superior in females versus males. Academic sites saw higher SVR rates than community sites and were more likely to prescribe on label. Overall discontinuation rate was ~1% (5/476), and occurred only in the 24 week duration group.